2 for 1: Targeting tumor-associated macrophages and cancer cells with a novel MERTK-targeting antibody-drug conjugate (ADC)

Abstract Number: 35317

Shugaku Takeda[1], PuiChi Lo[1], Daniel Schefer[1], Maryam Siddiqui[2], Anudishi Tyagi[2], Venkata Lokesh Battula[2], Abhishek Maiti[2], Sohail Tavazoie[1,3], Masoud Tavazoie[1], David Martin Darst[1], Isabel Kurth[1];
[1]Inspirna, Inc., New York, NY, [2]MD Anderson Cancer Center, Houston, TX, [3]The Rockefeller University, New York, NY

Introduction

- Our RNA-DRIVEr™ platform identified MERTK as a key driver of metastatic progression[1]
- MERTK is a member of the TAM family of receptor tyrosine kinases
- MERTK Is overexpressed in various solid and hematologic cancers and expression is associated with worse outcomes[2-11]
- Binding of its ligands (Gas6, protein S) drives cancer cell proliferation, survival, invasion and drug resistance[4, 12]
- MERTK is expressed on M2 tumor-associated macrophages (TAMs), playing a role in the production of immunosuppressive cytokines and efferocytosis[12]
- MERTK expression within retinal pigment epithelium (RPE) has resulted in retinal degeneration in animal models exposed to MERTK complete antagonists[13]

Background

RGX-019

- Humanized mAb with high affinity and specificity to human MERTK (Kd = 3.0 nM) with partial agonist activity[14]
- Efficiently internalizes MERTK leading to degradation of surface MERTK
- As a partial agonist, blocks binding of Gas6 ligand and diminishes signal transduction
- Induces expression of pro-inflammatory cytokines in M2 macrophages

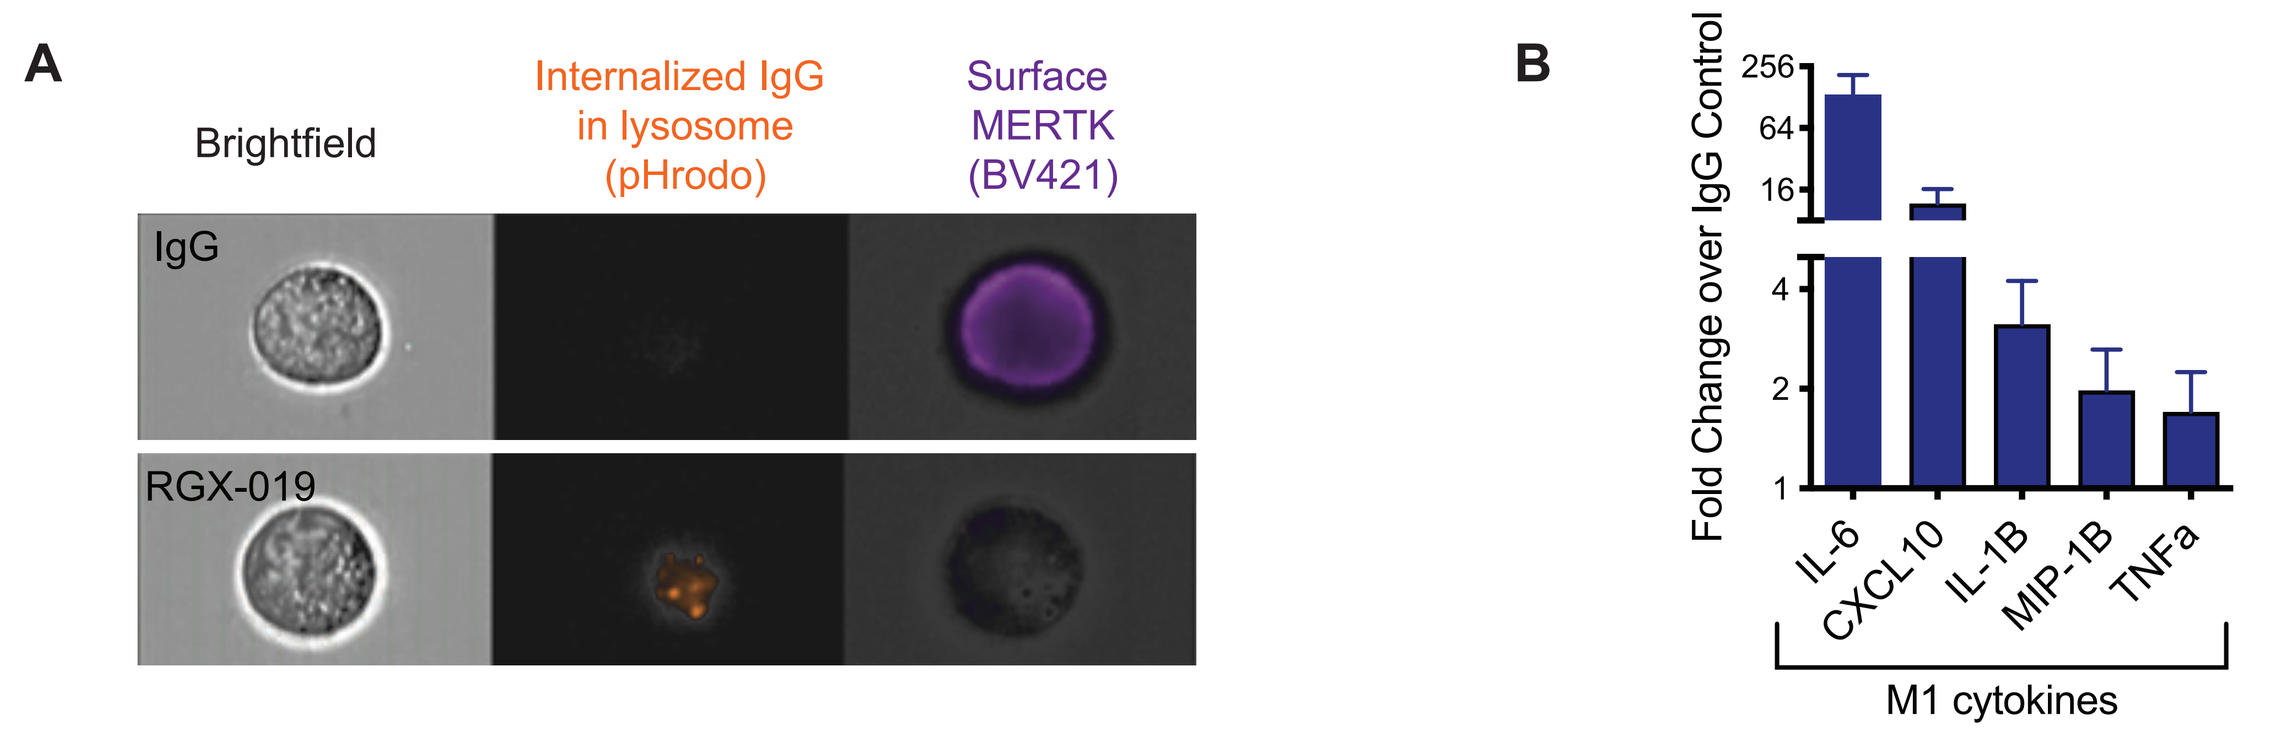
A
Brightfield
Internalized IgG in lysosome (pHrodo)
Surface MERTK (BV421)
IgG
RGX-019
B
Fold Change over IgG Control
IL-6
CXCL10
IL-1B
MIP-1B
TNFa
M1 cytokines

Figure 1. (A) RGX-019 Internalization. RGX-019 and IgG control were labeled with a pH-sensitive dye, pHrodo, that becomes fluorescent in low pH (eg. lysosomes).SK-Mel5 melanoma cells were treated with 6.7 nM pHrodo-labeled RGX-019 or IgG control for 24 hr before imaging with Image-Stream flow cytometer. **(B) Human cytokine array analysis.** M2 macrophages were differentiated from human monocytes by culturing with M-CSF for 7 days, and treated with 6.7 nM RGX-019 or IgG for 48hr. n=4; mean +/- SEM.

RGX-019-MMAE

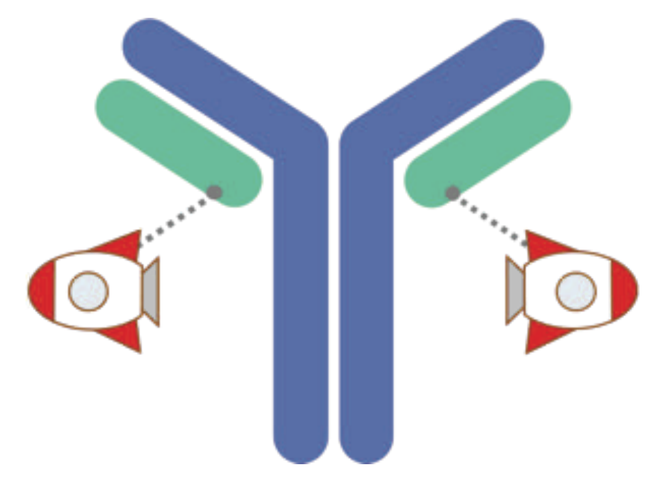

- RGX-019 MMAE-conjugated ADC
- MMAE payload: direct killing of mitotic cells & bystander efficacy
- Site-specific conjugation
- DAR 4
- Cleavable linker
- Additional spacer moiety to increase solubility

2 for 1 approach

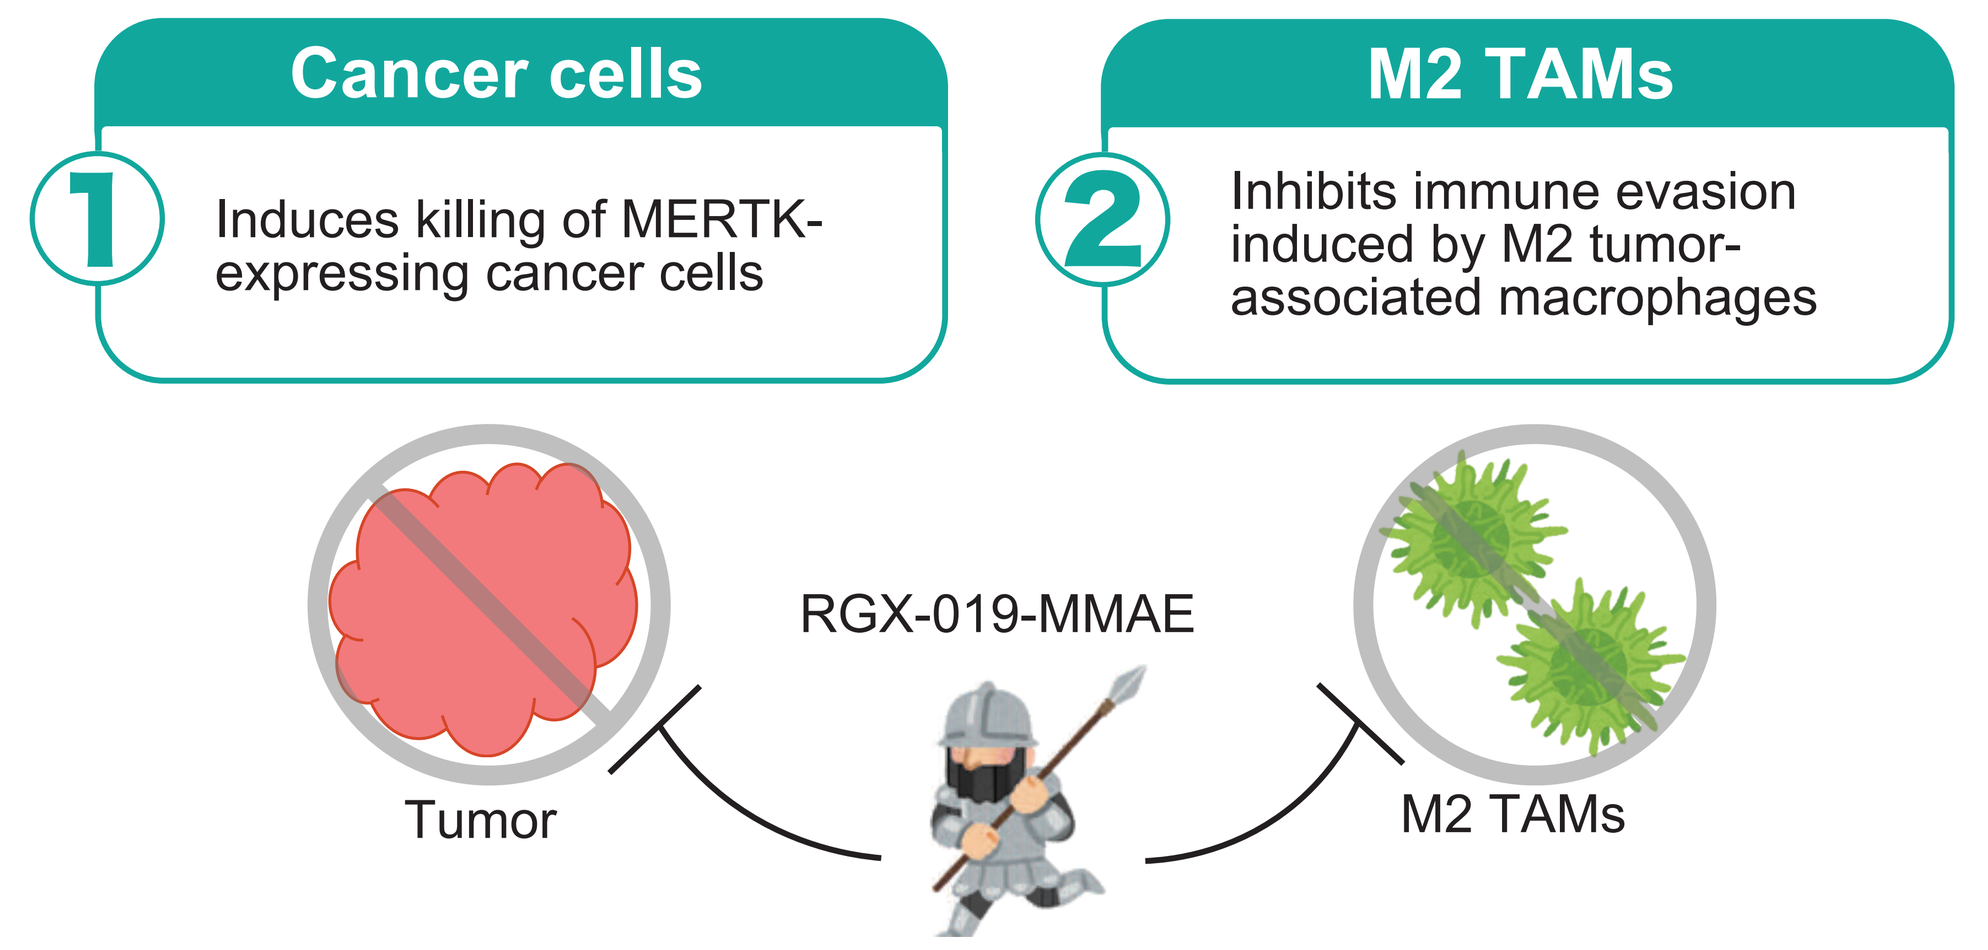
Cancer cells
1 Induces killing of MERTK-expressing cancer cells
M2 TAMs
2 Inhibits immune evasion induced by M2 tumor-associated macrophages
Tumor
RGX-019-MMAE
M2 TAMs

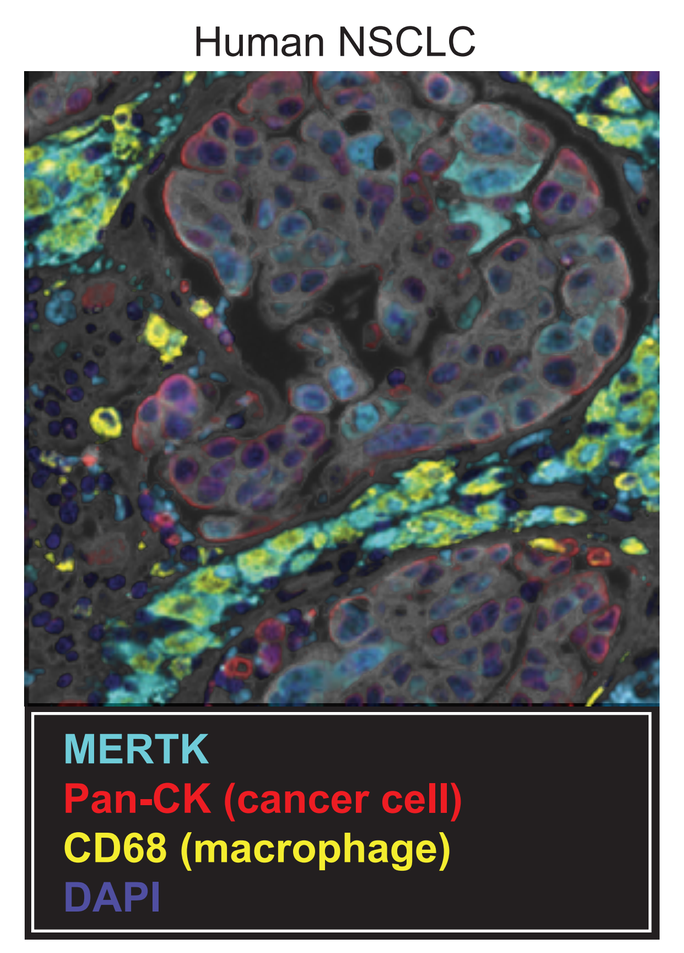
Human NSCLC
MERTK
Pan-CK (cancer cell)
CD68 (macrophage)
DAPI

Results

MERTK expression is observed in human solid cancers and AML

Expression of MERTK is seen in the majority of ovarian, urothelial, pancreas, and non-small cell lung cancers (NSCLC)

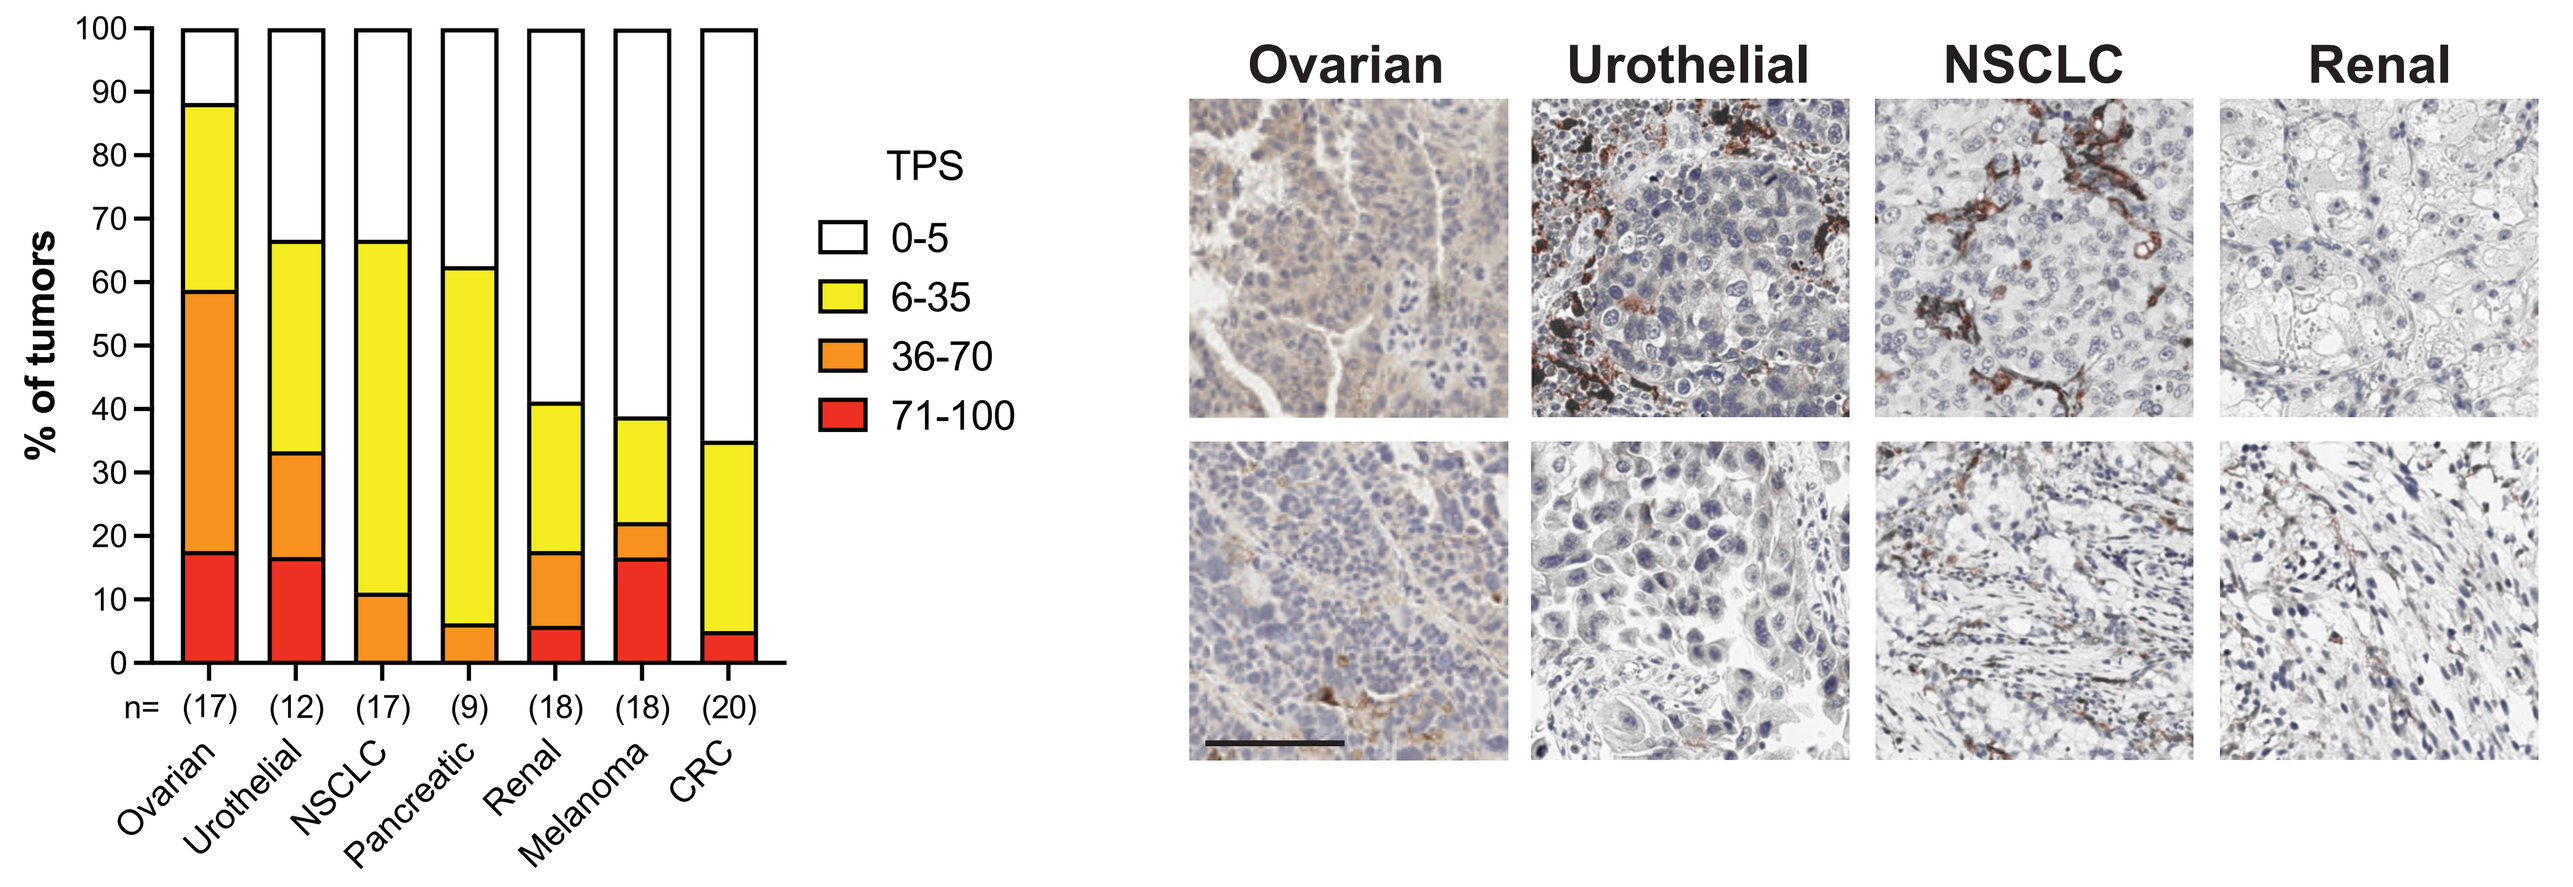
% of tumors
TPS
0-5
6-35
36-70
71-100
n= (17) (12) (17) (9) (18) (18) (20)
Ovarian
Urothelial
NSCLC
Pancreatic
Renal
Melanoma
CRC
Ovarian
Urothelial
NSCLC
Renal

Figure 2. MERTK immunohistochemistry (IHC) in human tumors. MERTK expression in human tumors across seven indications was analyzed by IHC and quantified as Tumor Proportion Score (TPS). Scale bar 100 µm.

Higher MERTK expression is found in M3 and M5 subtypes of AML as well as AML with t(9;11) translocations or PTPN11 mutations

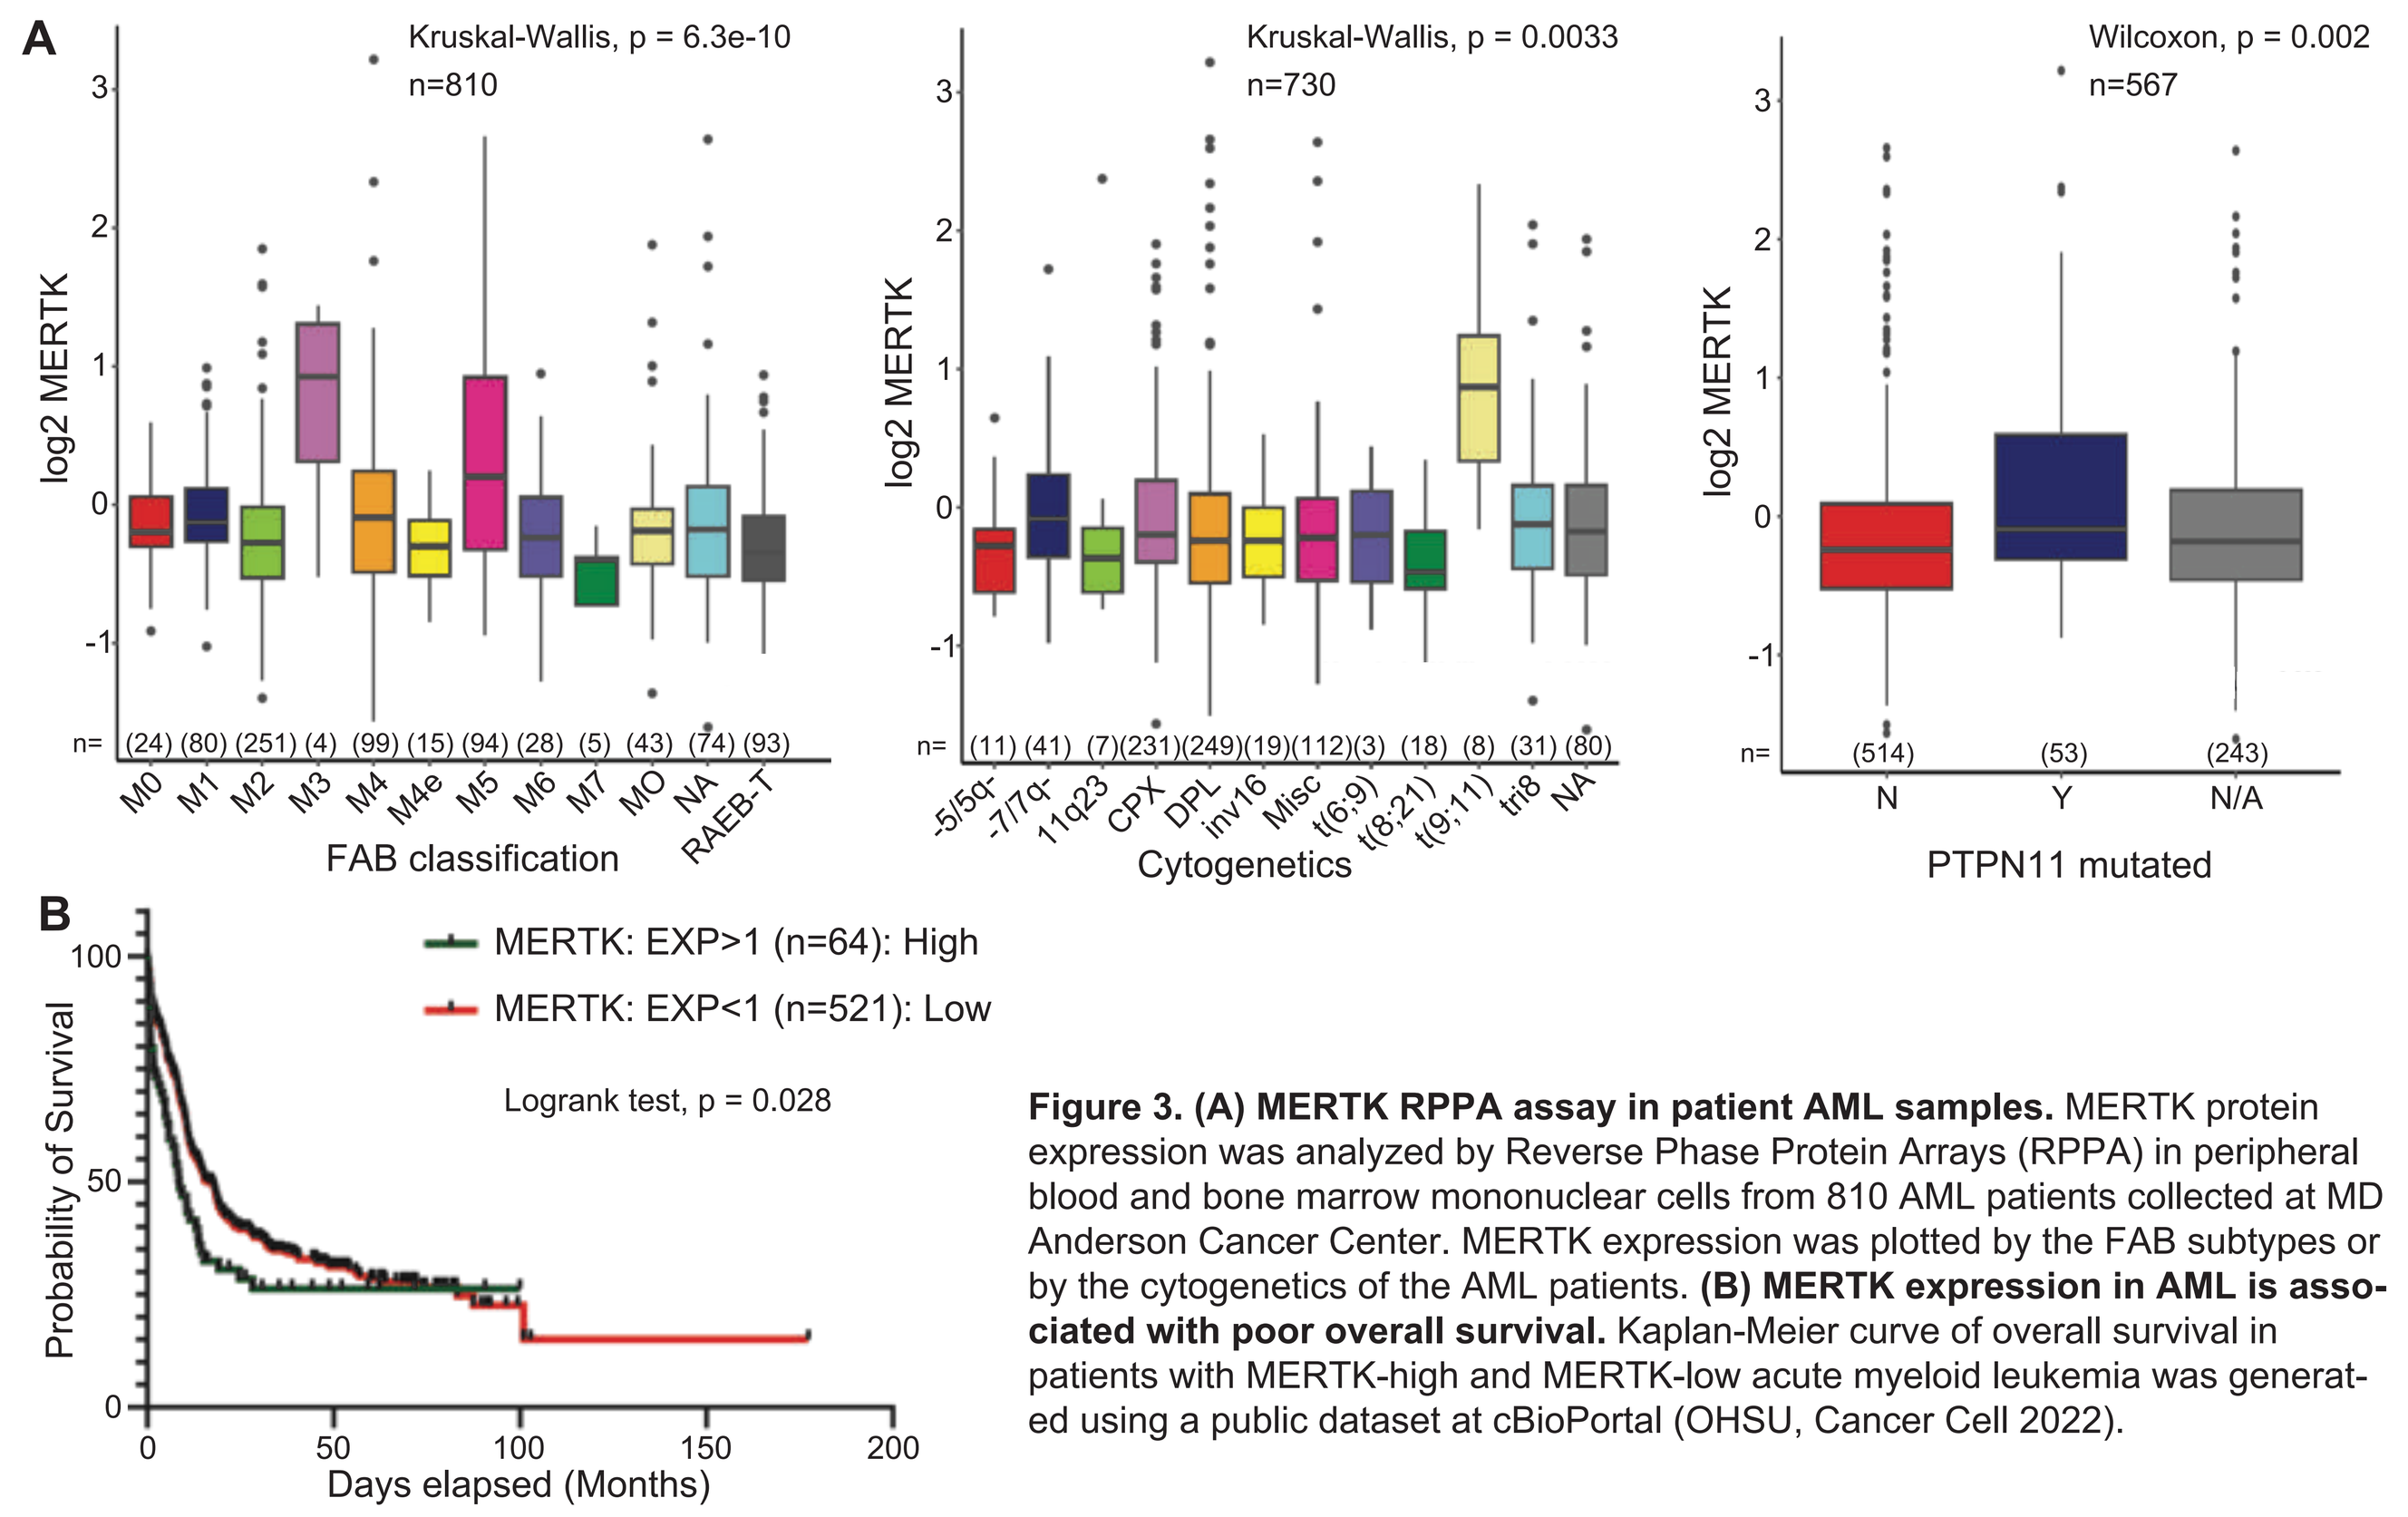
A
Kruskal-Wallis, p = 6.3e-10
n=810
log2 MERTK
FAB classification
Kruskal-Wallis, p = 0.0033
n=730
Cytogenetics
Wilcoxon, p = 0.002
n=567
PTPN11 mutated
N
Y
N/A
B
MERTK: EXP>1 (n=64): High
MERTK: EXP<1 (n=521): Low
Logrank test, p = 0.028
Probability of Survival
Days elapsed (Months)

Figure 3. (A) MERTK RPPA assay in patient AML samples. MERTK protein expression was analyzed by Reverse Phase Protein Arrays (RPPA) in peripheral blood and bone marrow mononuclear cells from 810 AML patients collected at MD Anderson Cancer Center. MERTK expression was plotted by the FAB subtypes or by the cytogenetics of the AML patients. **(B) MERTK expression in AML is associated with poor overall survival.** Kaplan-Meier curve of overall survival in patients with MERTK-high and MERTK-low acute myeloid leukemia was generated using a public dataset at cBioPortal (OHSU, Cancer Cell 2022).

RGX-019-MMAE demonstrates efficient internalization and target-dependent in vitro cytotoxicity

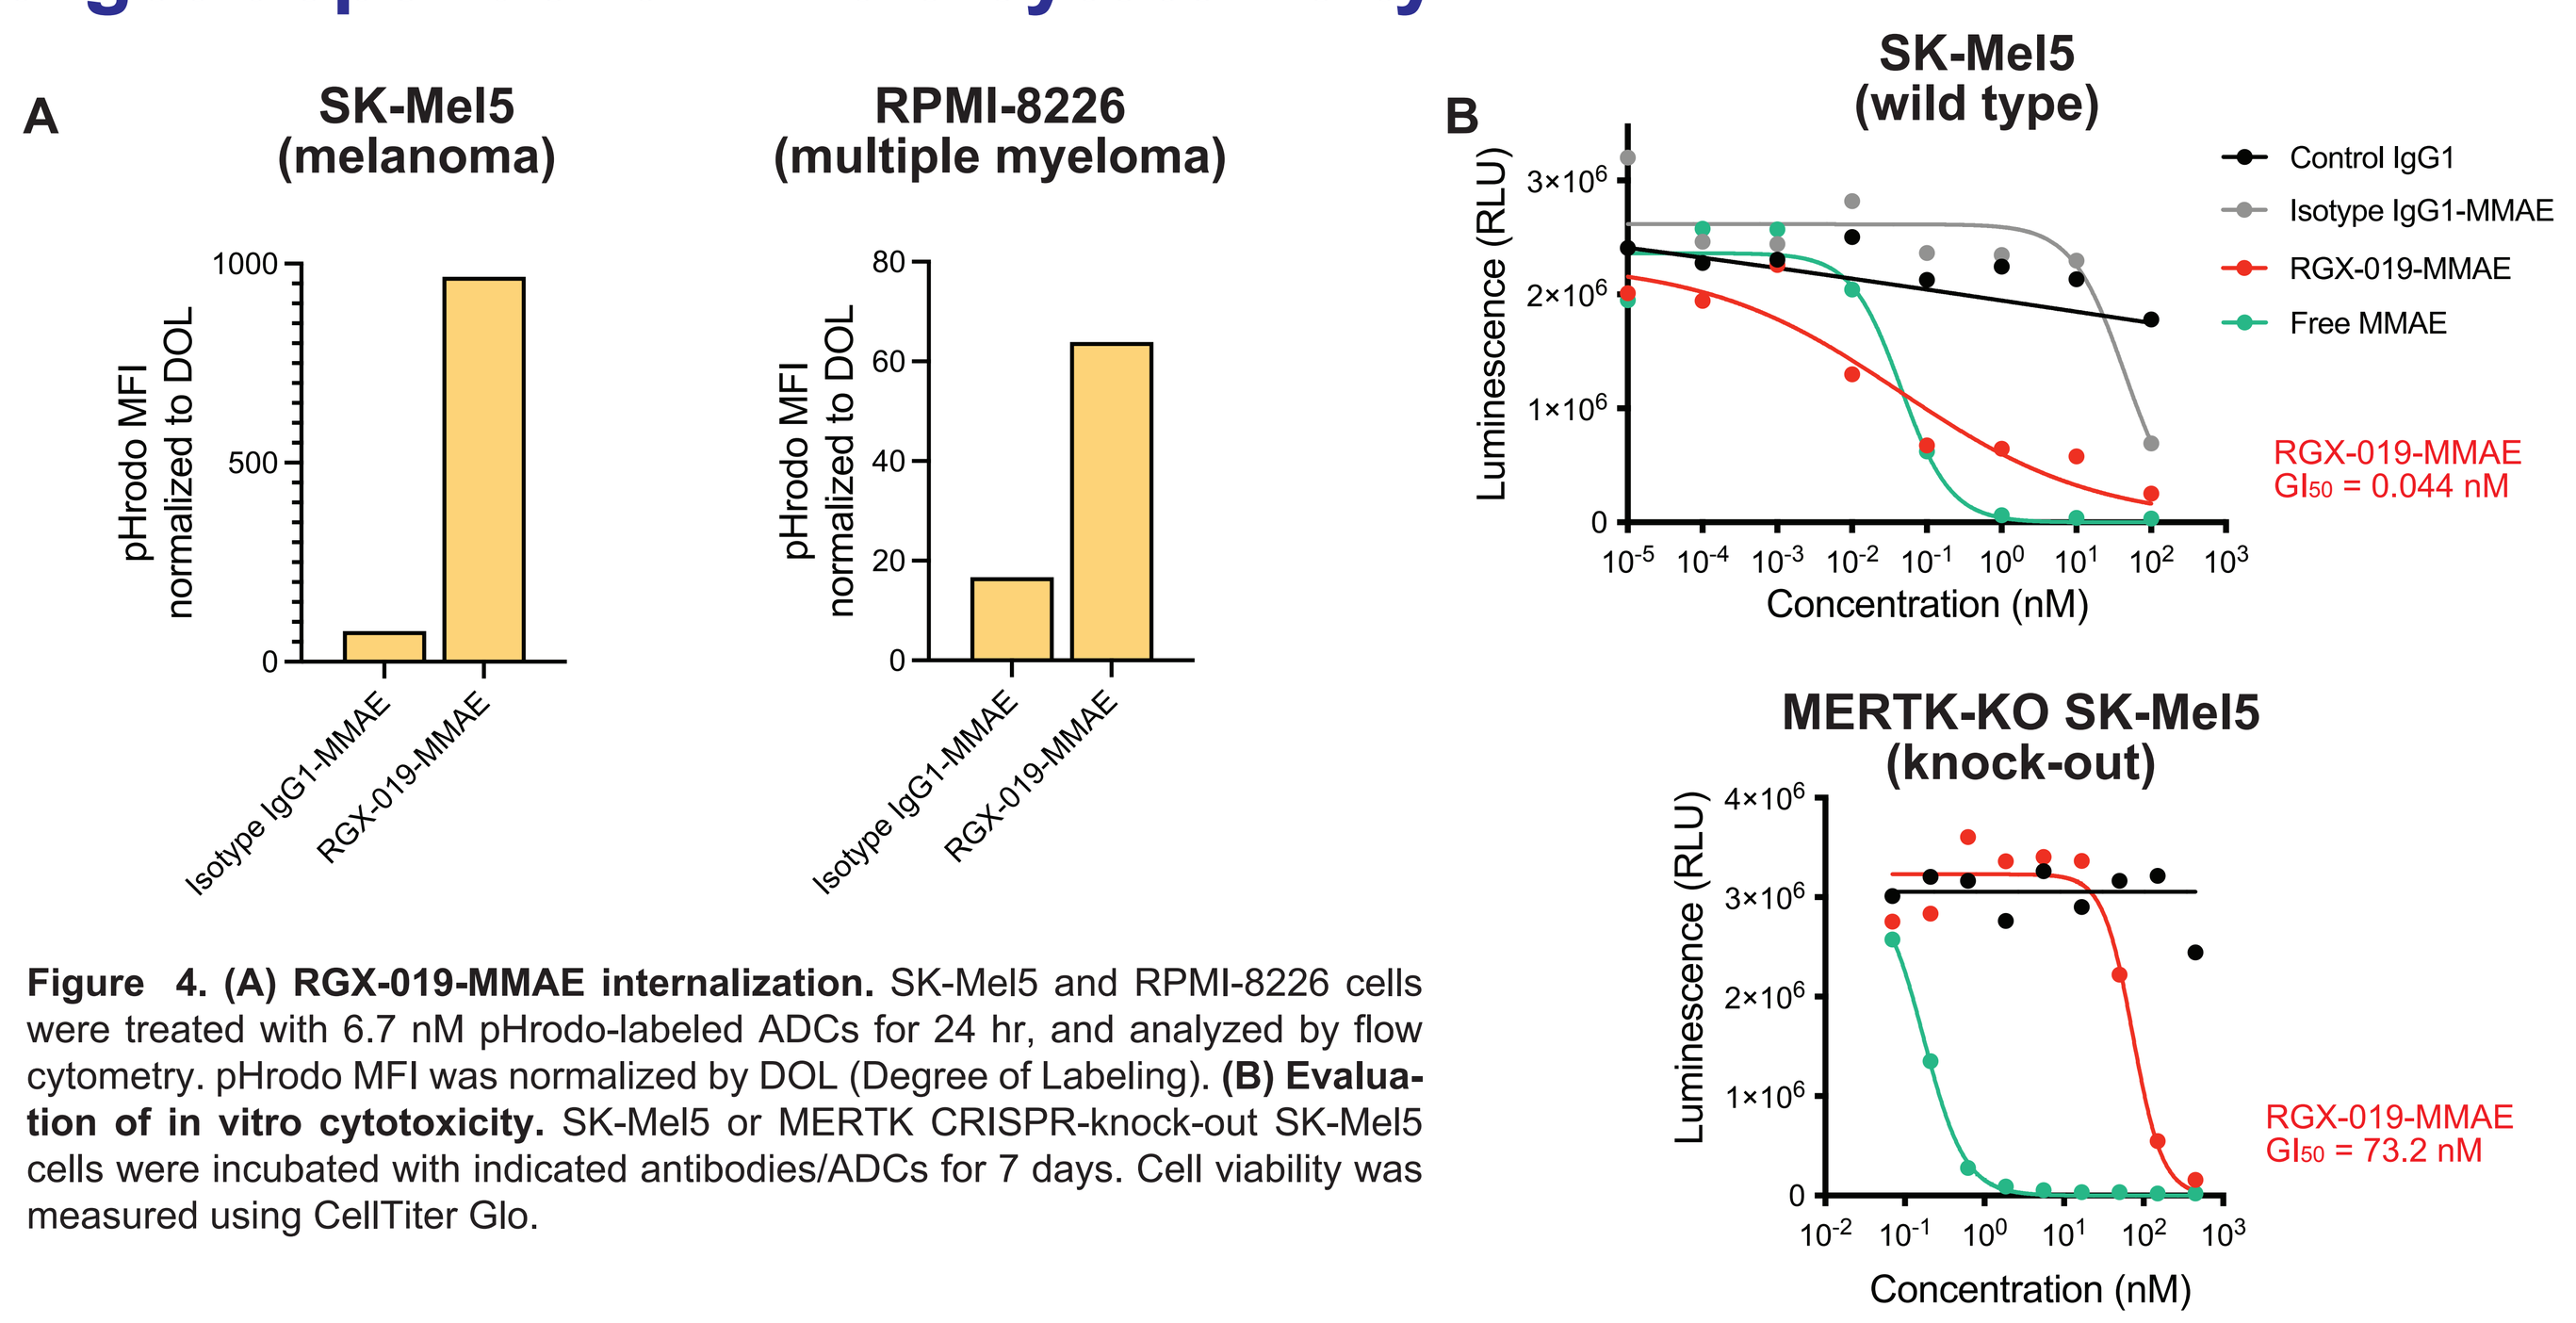
A
SK-Mel5 (melanoma)
RPMI-8226 (multiple myeloma)
pHrodo MFI normalized to DOL
Isotype IgG1-MMAE
RGX-019-MMAE
B
SK-Mel5 (wild type)
Control IgG1
Isotype IgG1-MMAE
RGX-019-MMAE
Free MMAE
RGX-019-MMAE GI50 = 0.044 nM
Luminescence (RLU)
Concentration (nM)
MERTK-KO SK-Mel5 (knock-out)
RGX-019-MMAE GI50 = 73.2 nM

Figure 4. (A) RGX-019-MMAE internalization. SK-Mel5 and RPMI-8226 cells were treated with 6.7 nM pHrodo-labeled ADCs for 24 hr, and analyzed by flow cytometry. pHrodo MFI was normalized by DOL (Degree of Labeling). **(B) Evaluation of in vitro cytotoxicity.** SK-Mel5 or MERTK CRISPR-knock-out SK-Mel5 cells were incubated with indicated antibodies/ADCs for 7 days. Cell viability was measured using CellTiter Glo.

RGX-019-MMAE exhibits robust in vivo anti-tumor efficacy

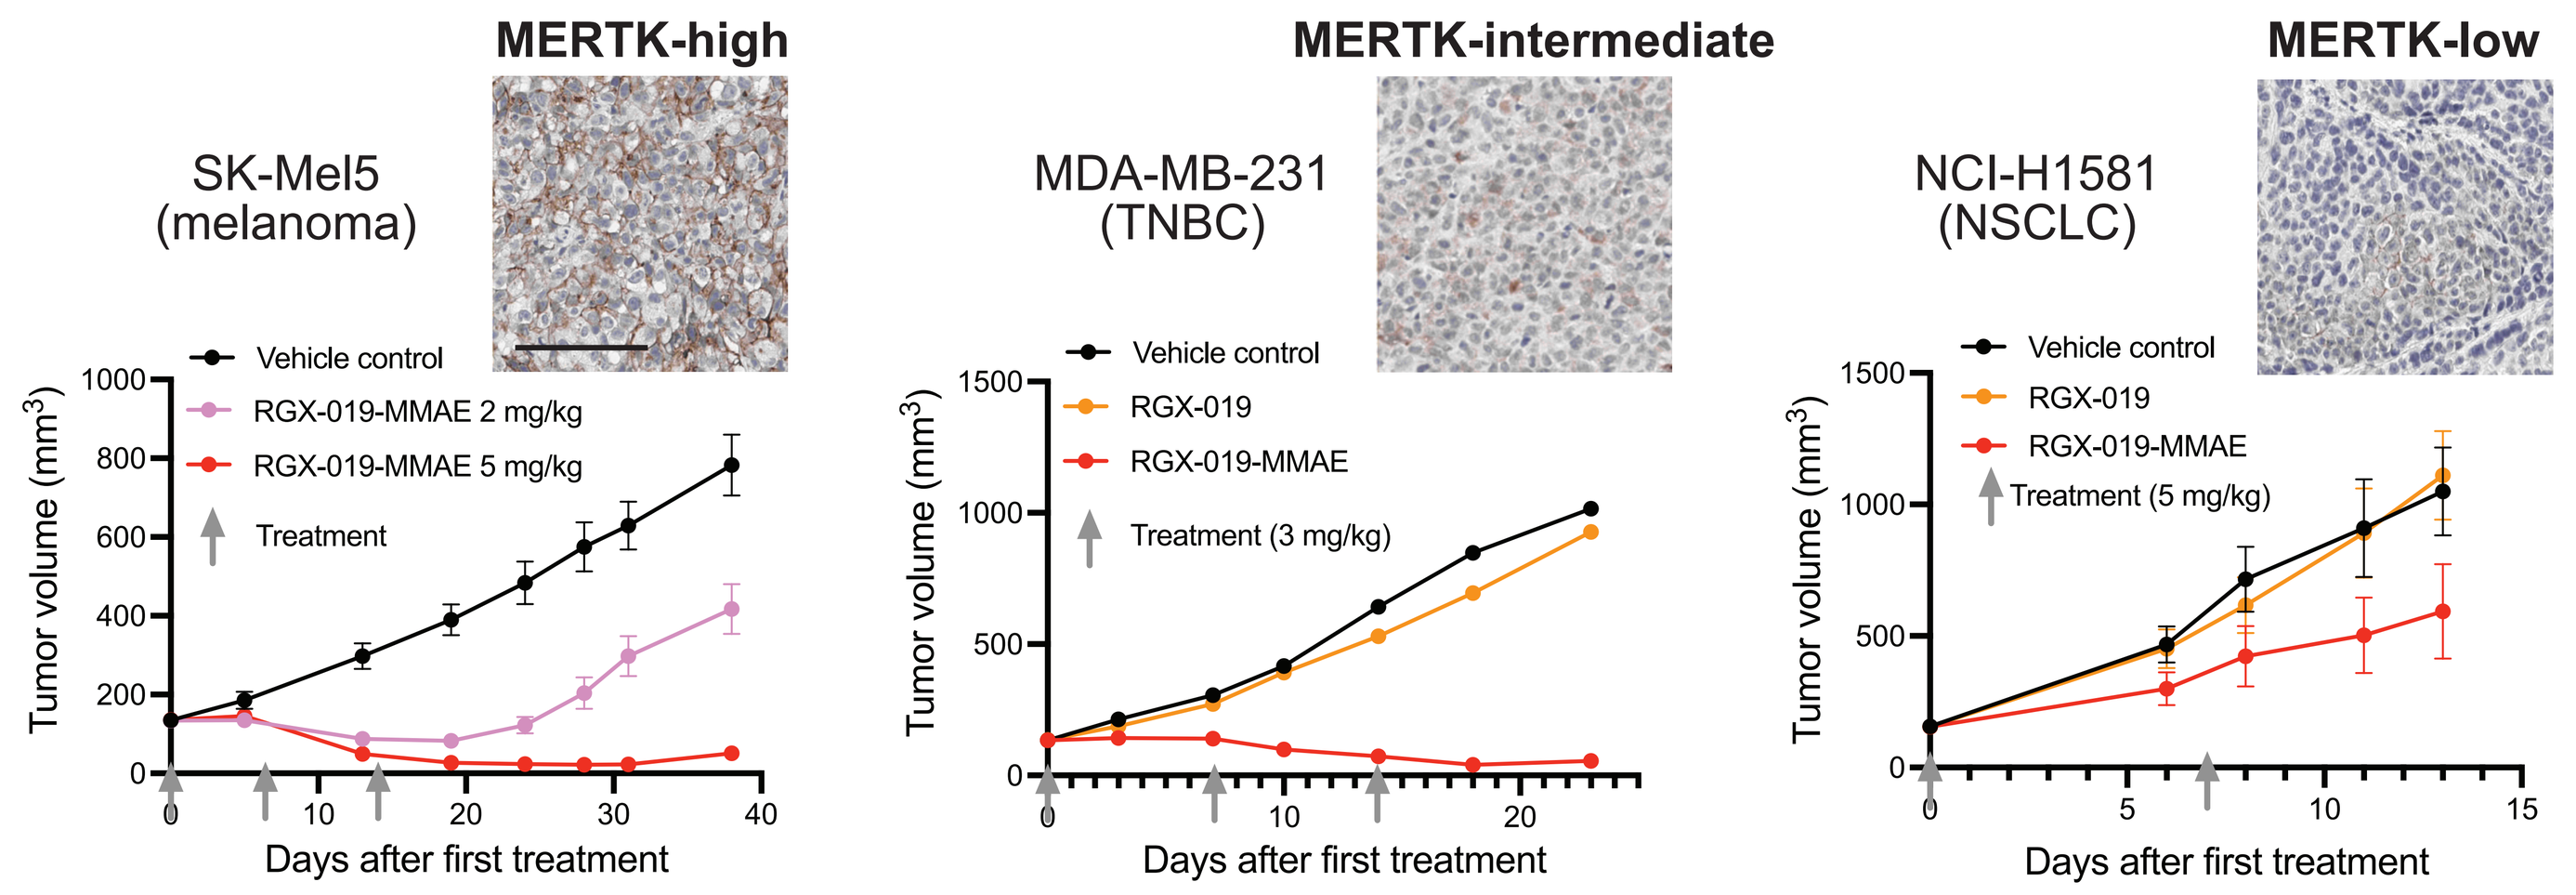
MERTK-high
SK-Mel5 (melanoma)
Vehicle control
RGX-019-MMAE 2 mg/kg
RGX-019-MMAE 5 mg/kg
Treatment
MERTK-intermediate
MDA-MB-231 (TNBC)
RGX-019
RGX-019-MMAE
Treatment (3 mg/kg)
MERTK-low
NCI-H1581 (NSCLC)
Treatment (5 mg/kg)
Tumor volume (mm3)
Days after first treatment

Figure 5. In vivo anti-tumor efficacy in xenograft models. SK-Mel5 or NCI-H1581 cells were injected subcutaneously into NSG or athymic mice, respectively. MDA-MB-231 cells were injected into the mammary fat pad of athymic nude mice. n=8-10, mean +/- SEM. Images represent MERTK IHC staining of control tumors. Scale bar 100 µm.

RGX-019-MMAE shows bystander activity via M2 macrophages

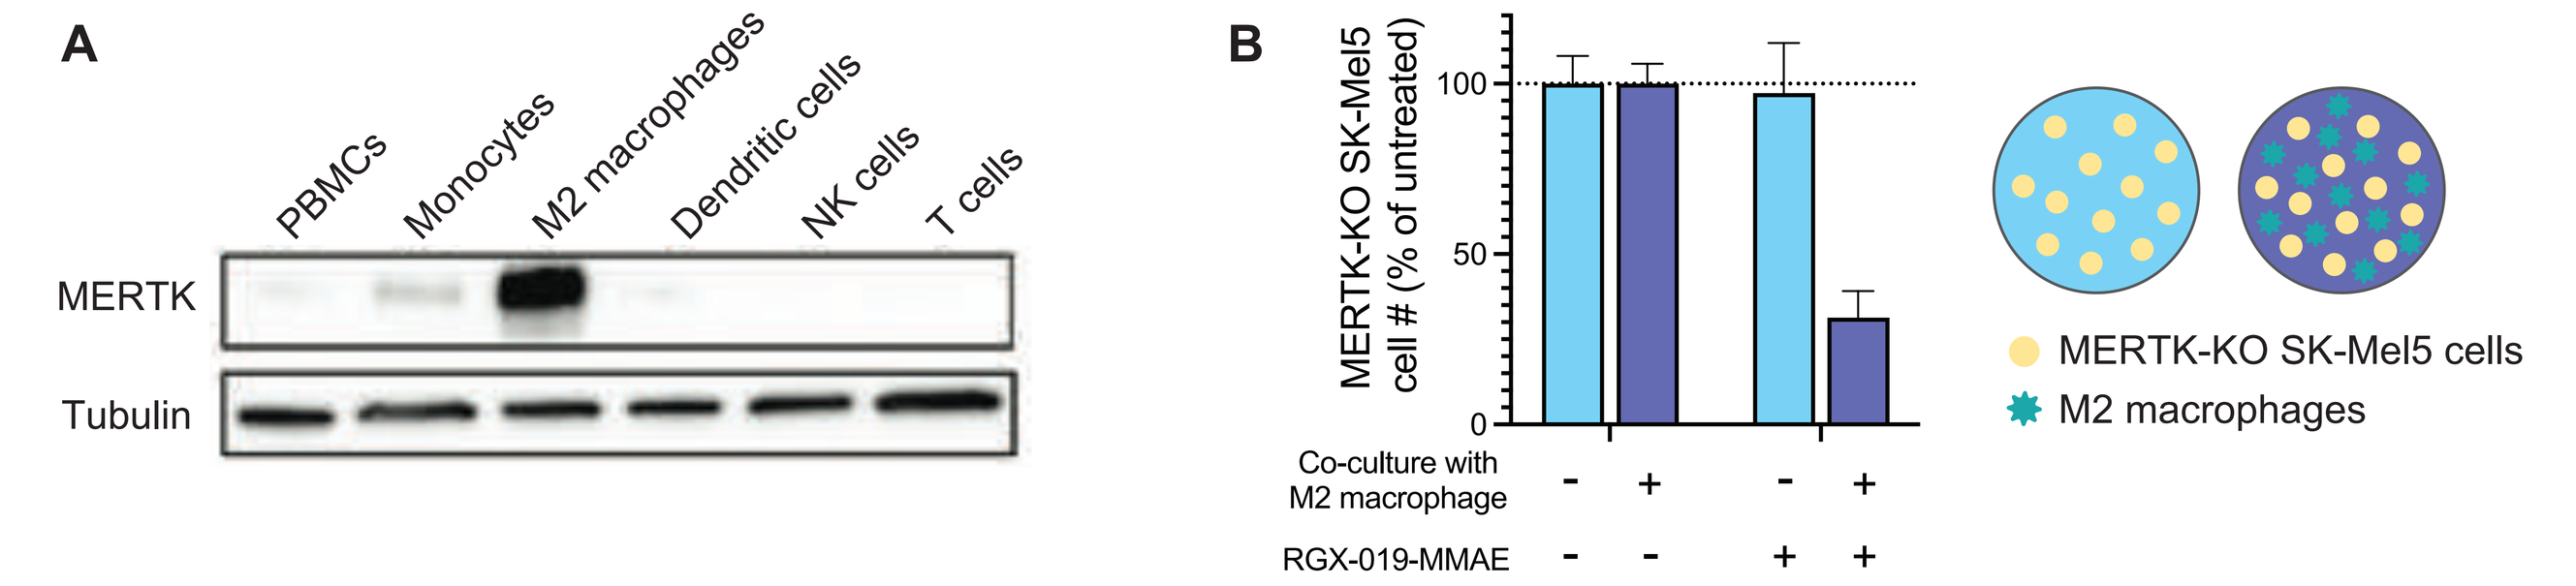
A
PBMCs
Monocytes
M2 macrophages
Dendritic cells
NK cells
T cells
MERTK
Tubulin
B
MERTK-KO SK-Mel5 cell # (% of untreated)
Co-culture with M2 macrophage
RGX-019-MMAE
MERTK-KO SK-Mel5 cells
M2 macrophages

Figure 6. (A) MERTK expression in human immune cells. In vitro differentiated M2 macrophages and other immune cells isolated from human PBMCs were analyzed by anti-MERTK Western blot. **(B) Evaluation of bystander-killing by M2 macrophages.** M2 macrophages were co-cultured with CFSE-labeled MERTK-KO SK-Mel5 cells in the presence of RGX-019-MMAE for 6 days. Cells were stained with anti-CD14 and anti-MERTK antibodies, and numbers of MERTK-KO SK-Mel5 cells were quantified by flow cytometry. n=3-4; mean +/- SD.

RGX-019-MMAE exhibits anti-tumor immune modulation

To address the contribution to tumor inhibition from the host immune system, we injected MMAE resistant tumor cells (MC38-HM) into either wild type mice or mice that express a humanized version of MERTK

- **Tumor growth was suppressed only in animals expressing humanized MERTK**
- **RGX-019-MMAE depletes tumoral M2 macrophages**

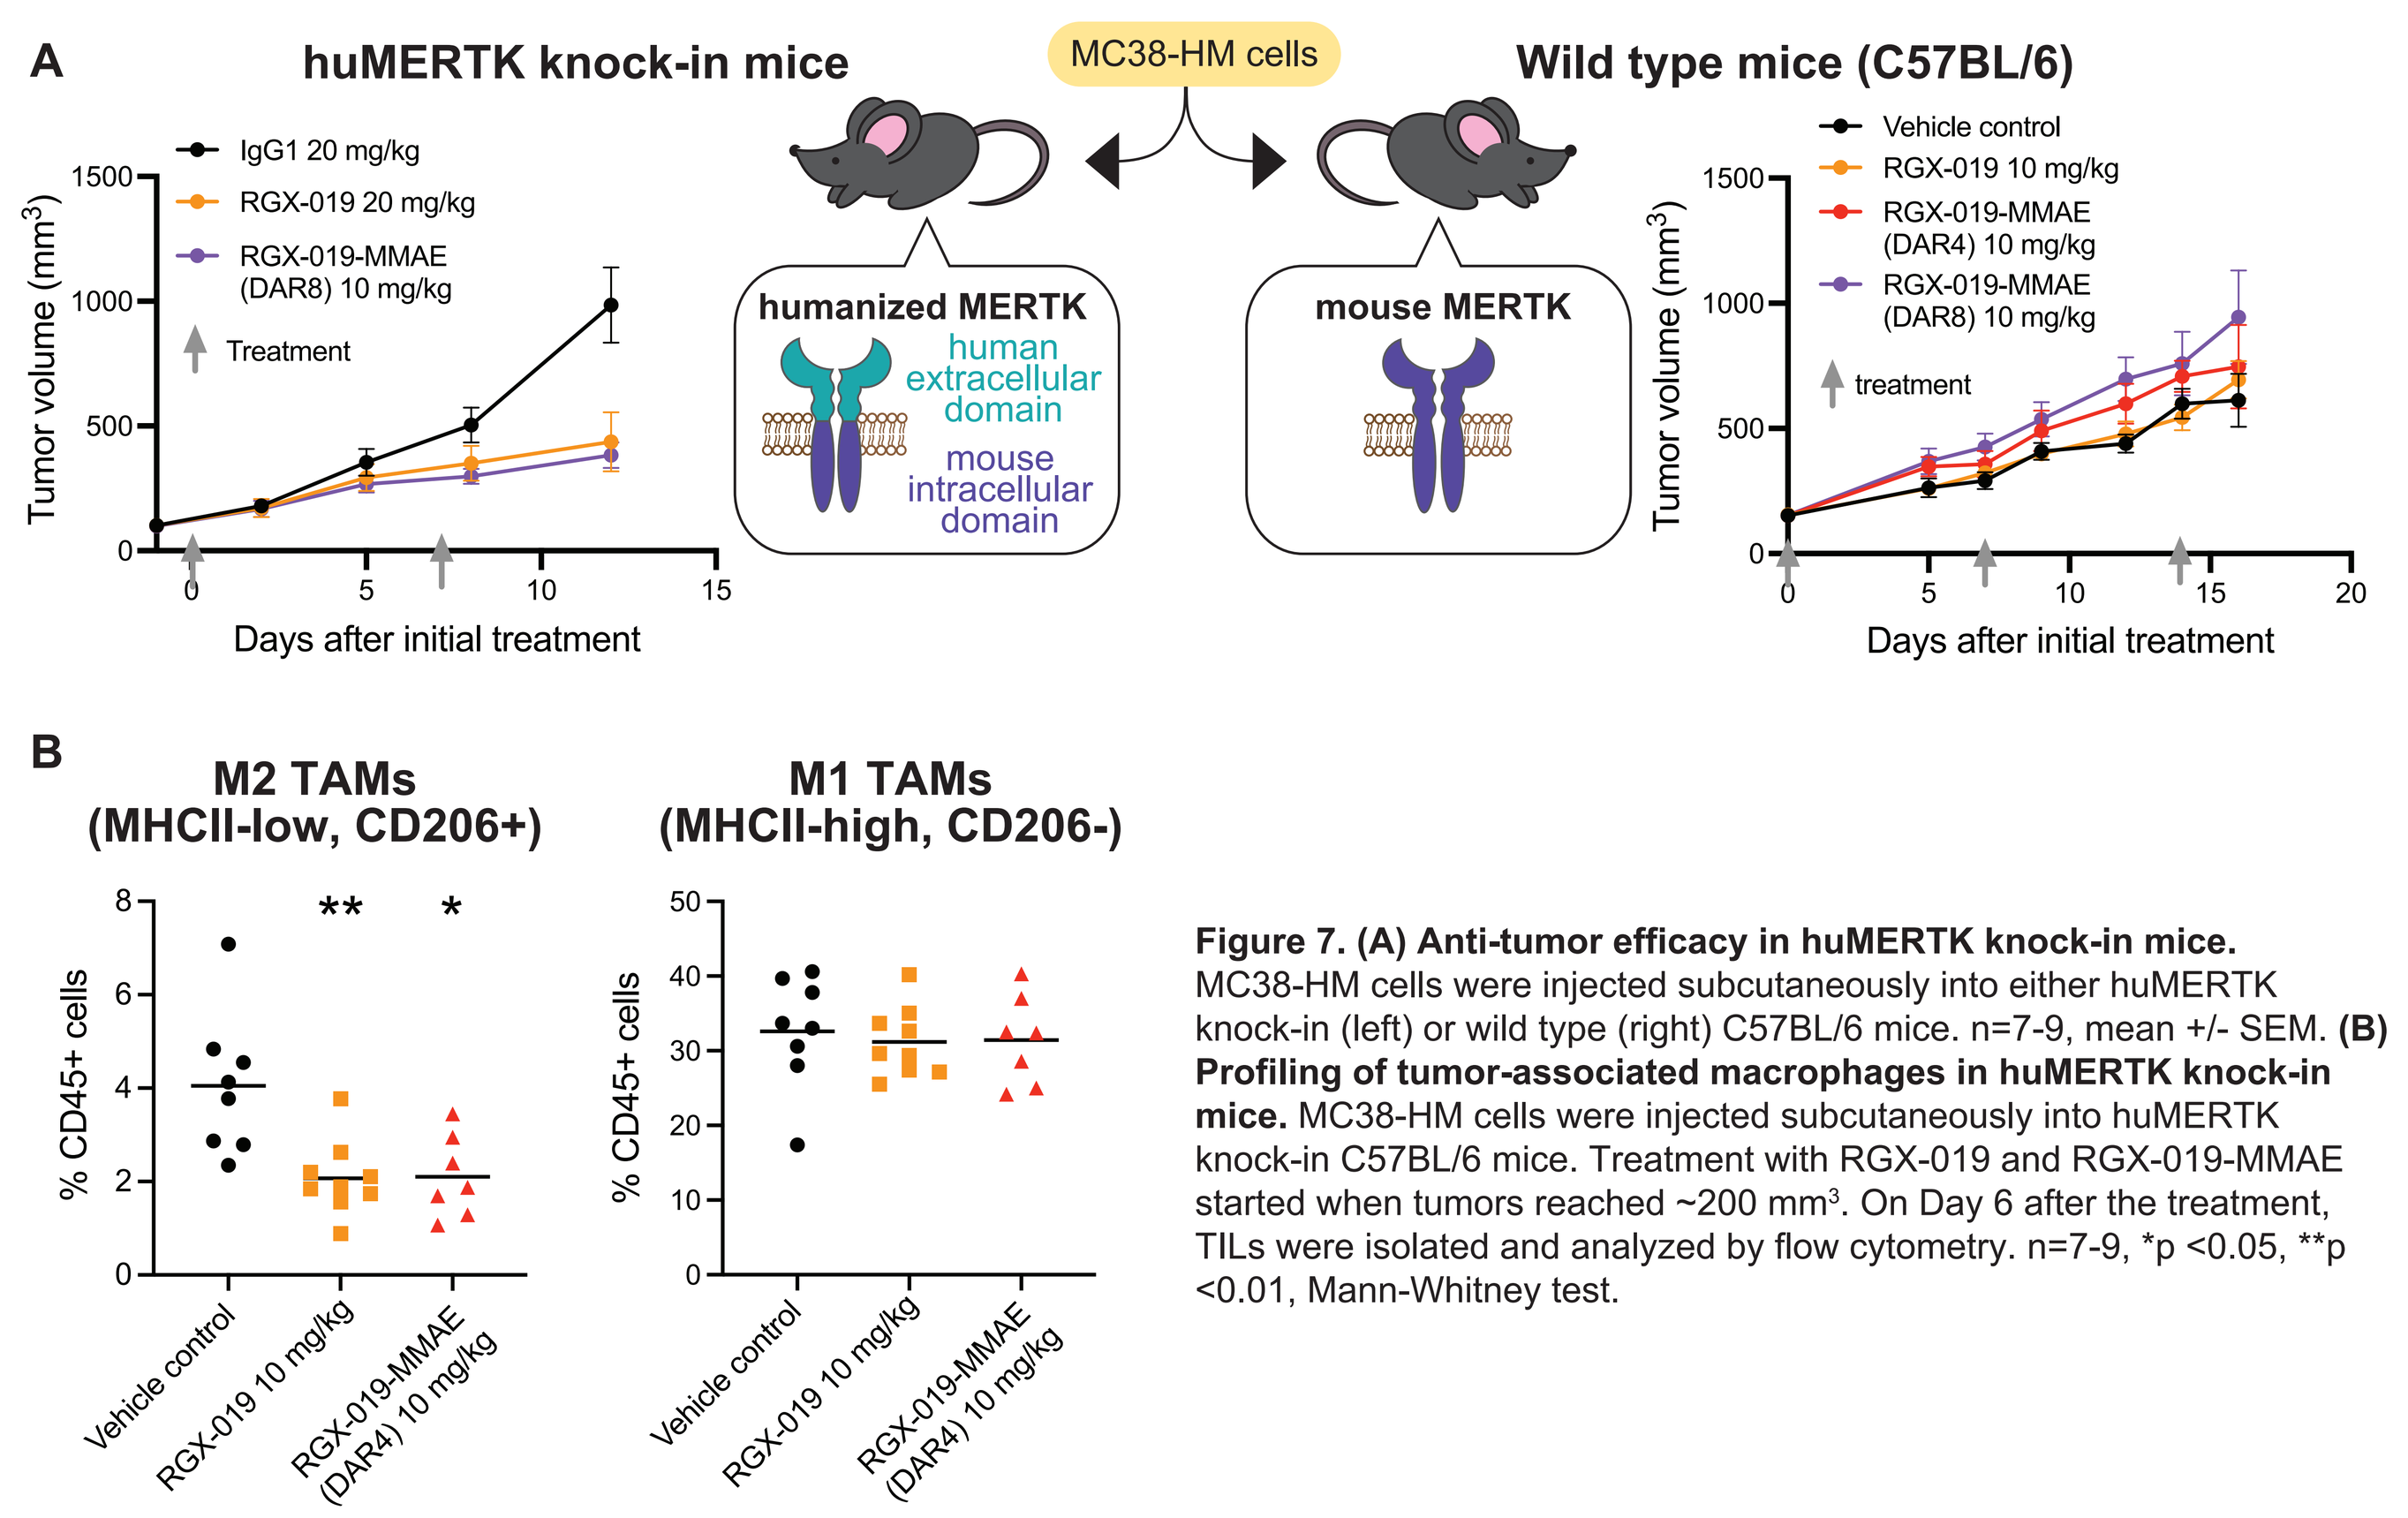
A
huMERTK knock-in mice
MC38-HM cells
Wild type mice (C57BL/6)
IgG1 20 mg/kg
RGX-019 20 mg/kg
RGX-019-MMAE (DAR8) 10 mg/kg
Treatment
humanized MERTK
human extracellular domain
mouse intracellular domain
mouse MERTK
Vehicle control
RGX-019 10 mg/kg
RGX-019-MMAE (DAR4) 10 mg/kg
RGX-019-MMAE (DAR8) 10 mg/kg
treatment
Tumor volume (mm3)
Days after initial treatment
B
M2 TAMs (MHCII-low, CD206+)
M1 TAMs (MHCII-high, CD206-)
% CD45+ cells
Vehicle control
RGX-019 10 mg/kg
RGX-019-MMAE (DAR4) 10 mg/kg

Figure 7. (A) Anti-tumor efficacy in huMERTK knock-in mice. MC38-HM cells were injected subcutaneously into either huMERTK knock-in (left) or wild type (right) C57BL/6 mice. n=7-9, mean +/- SEM. **(B) Profiling of tumor-associated macrophages in huMERTK knock-in mice.** MC38-HM cells were injected subcutaneously into huMERTK knock-in C57BL/6 mice. Treatment with RGX-019 and RGX-019-MMAE started when tumors reached ~200 mm³. On Day 6 after the treatment, TILs were isolated and analyzed by flow cytometry. n=7-9, *p <0.05, **p <0.01, Mann-Whitney test.

RGX-019-MMAE treatment is not associated with retinal degeneration in mice

Partial agonist activity of RGX-019 prevents complete inhibition of RPE phagocytosis, which is needed for maintenance of retinal function, as compared to a full MERTK inhibitor

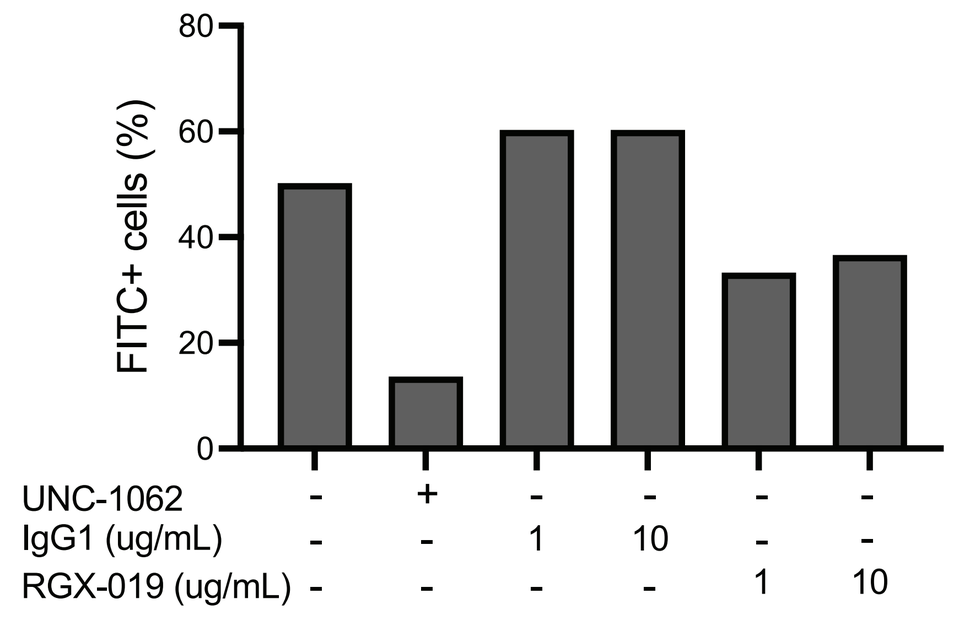
FITC+ cells (%)
UNC-1062
IgG1 (ug/mL)
RGX-019 (ug/mL)

Figure 8. Partial inhibition of phagocytosis by RPE cells. Primary human RPE (retinal pigmented epithelial) cells were differentiated in vitro. The cells were treated with IgG1, RGX-019 or a small molecule MERTK inhibitor UNC-1062 (10 uM) for 2 hr, then incubated with FITC-labeled bovine POS (photoreceptor outer segment) for 2 hr. The proportion of FITC-positive RPE cells was analyzed by flow cytometry.

RGX-019-MMAE treated mice have an intact retina

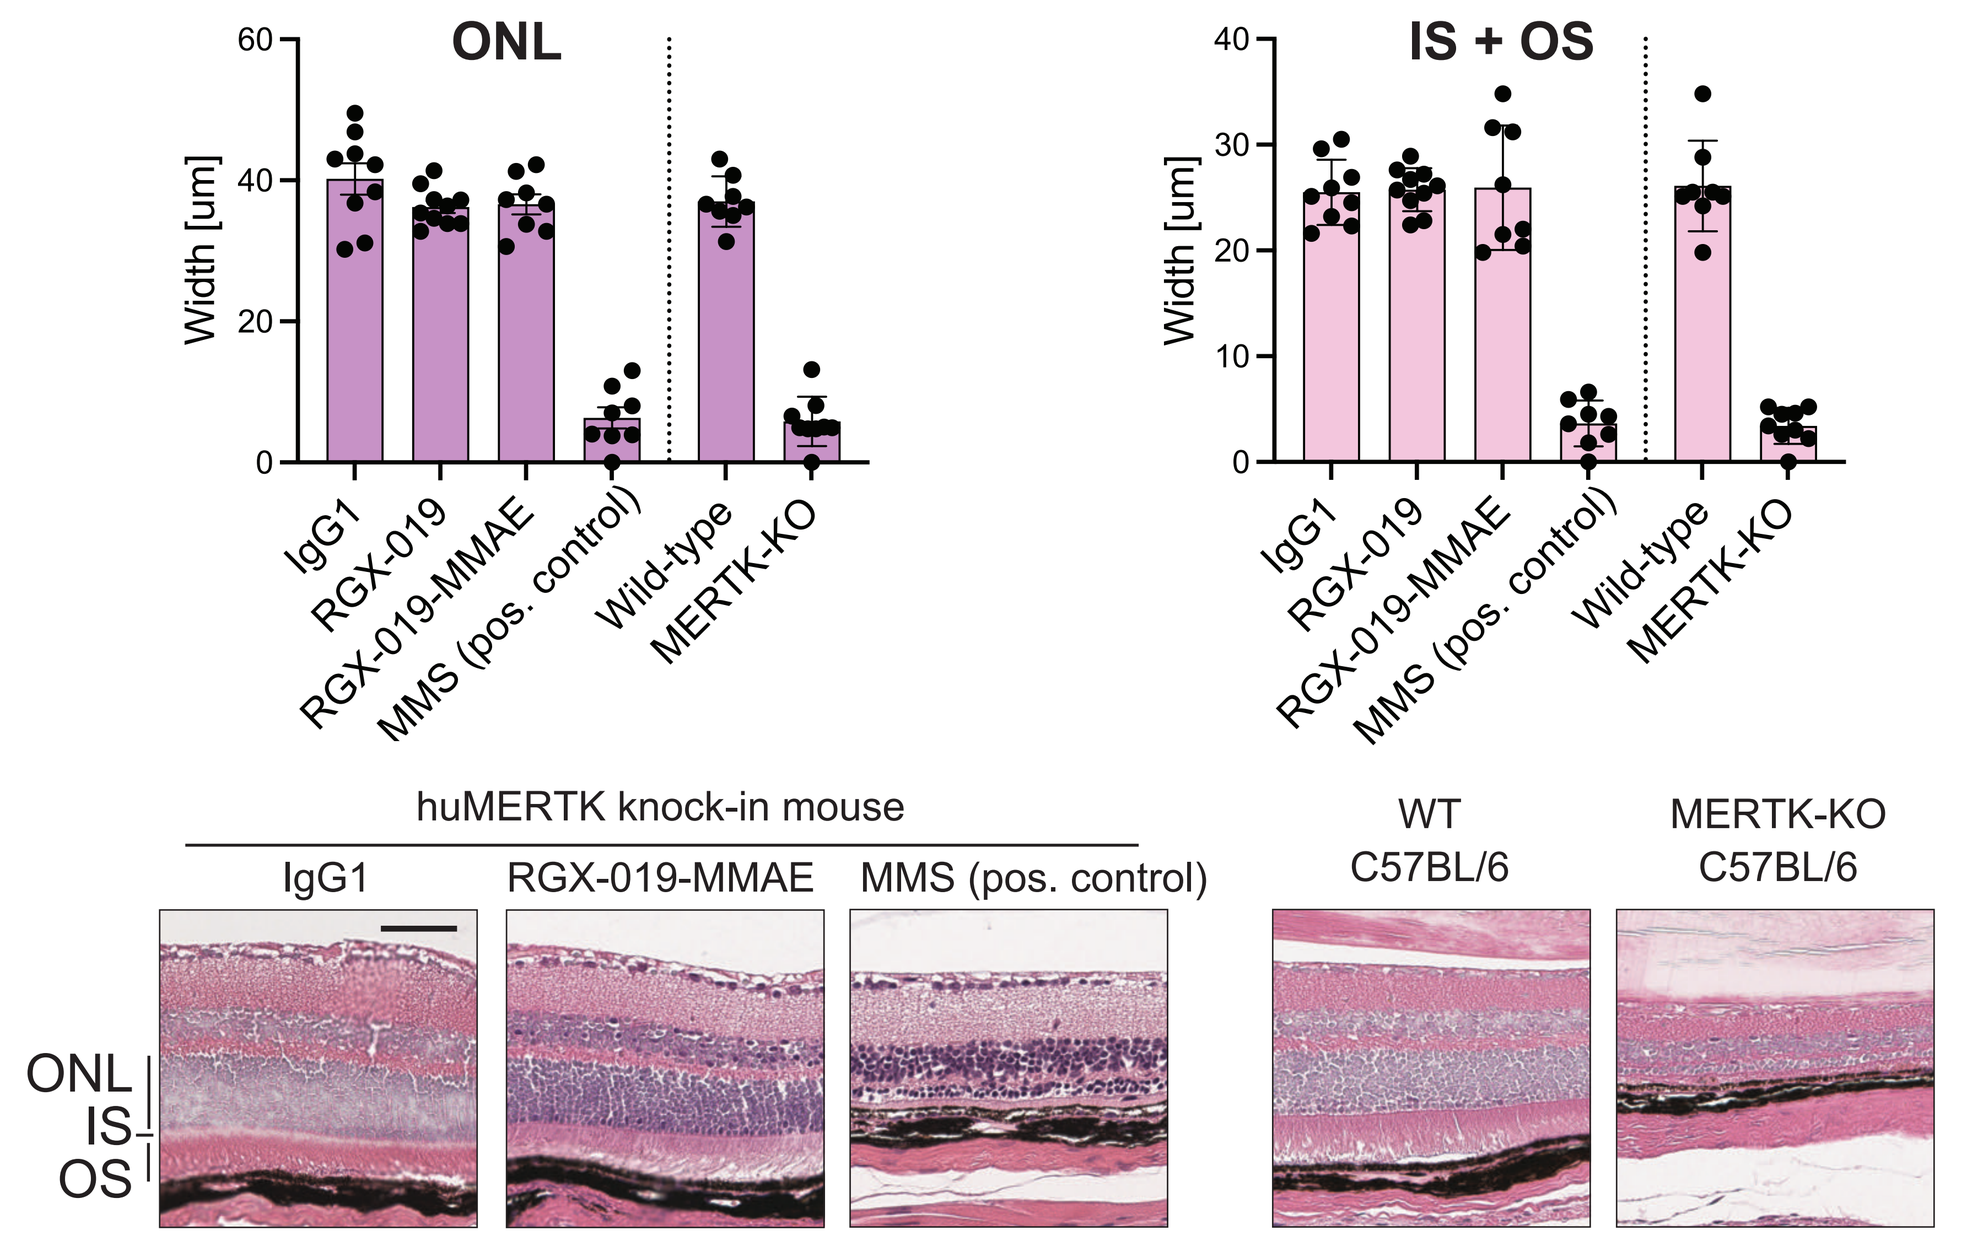
ONL
IS + OS
Width [um]
IgG1
RGX-019
RGX-019-MMAE
MMS (pos. control)
Wild-type
MERTK-KO
huMERTK knock-in mouse
WT C57BL/6
MERTK-KO C57BL/6
ONL
IS
OS

Figure 9. Assessment of retinal degeneration in mice. huMERTK-KI mice were treated with 5 mg/kg IgG1, RGX-019, or RGX-019-MMAE by weekly IV administration for 28 days and eyes were collected for H&E staining. As a positive control, an alkylating agent MMS (methyl methanesulfonate) was administered at 75 mg/kg by a single IP. The thickness of ONL (outer nuclear layer) and IS+OS (photoreceptor inner segment, outer segment) were quantified as measures of retinal degeneration (n=8-10). Scale bar = 50 µm.

Summary

- MERTK is overexpressed in multiple solid and hematologic cancers with expression in healthy tissues generally limited to the retinal pigment epithelium and immune cells, mainly macrophages
- RGX-019 triggers MERTK receptor internalization and partial agonist activity enabling selective delivery of toxic payload to MERTK expressing cancer cells
- Inspirna has developed an RGX-019 based MMAE ADC to enhance cancer cell killing and activate anti-tumor immunity
- Robust in vitro and in vivo efficacy of RGX-019-MMAE is seen in solid and hematologic cancers
- Lack of retinal degeneration observed in cynomolgus monkey[14] and mice expressing huMERTK, after treatment with RGX-019 and RGX-019-MMAE, differentiates RGX-019-MMAE from complete antagonist inhibitors of MERTK

Acknowledgements

The authors thank Inspirna's team members, Dr. Robert Busby, Dr. Usman Azam, Dr. Robert Wasserman, and Dr. Anne Assmus, for their comments and support. We gratefully acknowledge the contributions of our collaborators, Dr. Suresh Anaganti (L2P Research labs; MDA-MB-231 study), Dr. Yingbei Chen (IHC scoring), Histowiz (IHC) and Abzena.

References

[1]Png KJ, et al. Nature. 2011, [2]Koda Y. Anticancer Research. 2018, [3]Waizenegger JS. Leukemia. 2015, [4]Linger RM, et al. Oncogene. 2013, [5]Xie S, et al. Oncotarget. 2015, [6]Wu J. Neur Oncology. 2018, [7]Schlegel J, et al. J. Clin. Inv. 2013, [8]Wu YM, et al. Cancer Res. 2004, [9]Schmitz R, et al. Oncotarget. 2016, [10]Yi JH, et al. Oncotarget. 2017, [11]Evans CO. J Clin Endocrinol Metab. 2001, [12]Huelse J, et al. Pharmacol Ther. 2020, [13]Prasad D, et al. Mol Cell Neurosci. 2006, [14]Takeda S, et al. AACR Annual Meeting 2020