Abstract #9686

Poster # LB-133

Correlative analysis of pharmacokinetics and pharmacodynamics of RGX-104, a first-in-class Liver-X-Receptor (LXR) agonist, and clinical outcomes in patients with advanced solid tumors

RGENIX

Presented at: 2020 AACR

Monica Mita[1], Alan Mita[1], Erika Hamilton[2], Gerald S. Falchook[3], Michael Postow[4], Bartosz Chmielowski[5], Russell J. Schilder[6], James Strauss[7], Emerson Lim[8], Shubham Pant[9], Angela Jain[10], Oliver Rixe[11], Rebecca Redman[12], Kevin B. Kim[13], Tomislav Dragovich[14], R. Donald Harvey[15], Igor Puzanov[16], Nimisha Schneider[17], Renee Deehan[17], Tobi Guennel[17], Joe Lin[17], Sohail Tavazoie[18], Roger Waltzman[19], Eric Rowinsky[19], Michael Szarek[19], Subhasree Sridhar[19], Robert Busby[19], Narayan Lebaka[19], Celia Andreu[19], Isabel Kurth[19], David Darst[19], Masoud Tavazoie[19], Syed Raza[19], Robert Wasserman[19], Foster C. Gonsalves[19]

[1]Cedars-Sinai Medical Center, Los Angeles, CA; [2]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN; [3]Sarah Cannon Research Institute at HealthONE, Denver, CO; [4]Weill-Cornell Medical Center and Memorial Sloan Kettering Cancer Center, New York, NY; [5]University of California, Los Angeles, Los Angeles, CA; [6]Thomas Jefferson University, Philadelphia, PA; [7]Mary Crowley Cancer Research, Dallas, TX; [8]Columbia University Medical Center, New York, NY; [9]MD Anderson Cancer Center, Houston, TX; [10]Fox Chase Cancer Center, Philadelphia, PA; [11]University of New Mexico Cancer Center, Albuquerque, NM; [12]University of Louisville, Louisville, KY; [13]Sutter Health, San Francisco, CA; [14]Banner Health/MD Anderson Cancer Center, Gilbert, AZ; [15]Winship Cancer Institute of Emory University, Atlanta, GA; [16]Roswell Park Cancer Center, Buffalo, NY; [17]Precision for Medicine, MD; [18]Rockefeller University, New York, NY; [19]RGENIX Inc., New York, NY

A. INTRODUCTION

RGX-104 Induces Antitumor Immunity Via LXR/ApoE Activation

RGX-104-mediated LXR/ApoE activation depletes Myeloid-Derived Suppressor Cells (MDSCs) to induce a T cell response against tumors

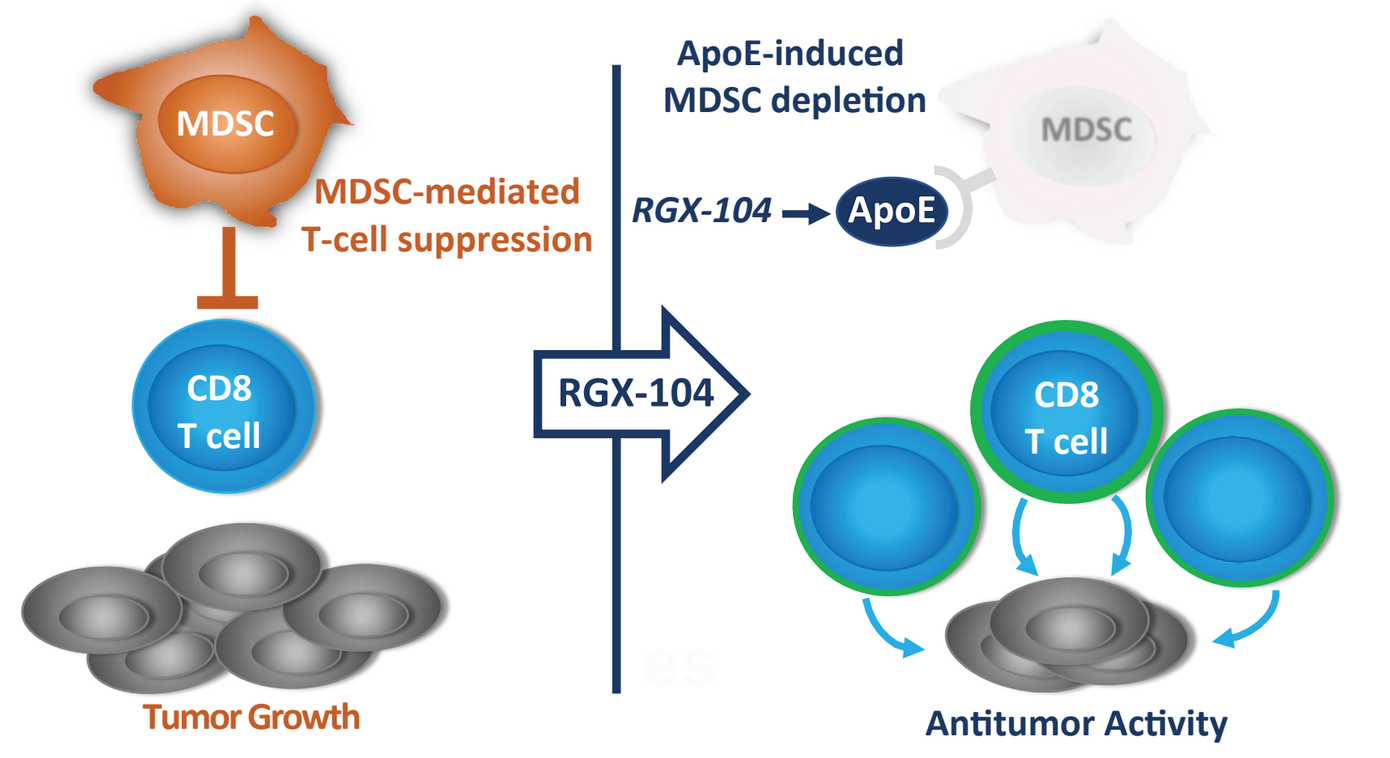
MDSC
MDSC-mediated T-cell suppression
CD8 T cell
Tumor Growth
RGX-104
ApoE-induced MDSC depletion
RGX-104 → ApoE
MDSC
CD8 T cell
Antitumor Activity

- Myeloid-derived suppressor cells (MDSCs) suppress antitumor immunity by inhibiting T cell activation and effector function
- The expansion of MDSCs in circulation and tumors of patients is associated with both innate and acquired resistance to checkpoint inhibitors (CPI) and chemotherapy, providing rationale to target MDSCs to reverse drug resistance and enhance activity of those agents (1)
- MDSC expansion, tumor angiogenesis, and poor clinical outcomes in patients as well as tumor-bearing mice is associated with reduction in ApoE protein levels (2,3)
- ApoE expression can be therapeutically activated by small-molecule agonists of the nuclear hormone receptor Liver-X Receptor (LXR)
- RGX-104 is a first-in-class oral small-molecule LXR agonist that activates ApoE expression, resulting in MDSC depletion, T cell activation, and antitumor activity as a single agent in murine models as well as in patients with cancer
- In addition to direct effects on MDSCs, RGX-104 modulates lipid metabolism, including induction of the cholesterol efflux transporters ABCA1 and ABCG1, which can be measured as pharmacodynamic markers of LXR activity (4)
- Here, we present pharmacodynamic data, including immune responses, from single agent and combination dose escalation studies of RGX-104 in patients with solid tumors - including CPI refractory patients - and correlate these pharmacodynamic effects with treatment outcomes

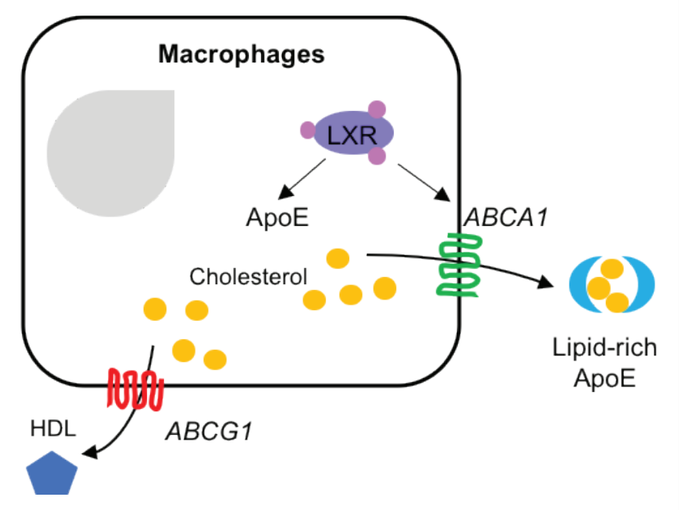
Macrophages
LXR
ApoE
ABCA1
Cholesterol
Lipid-rich ApoE
HDL
ABCG1

B. Methods

- Flow cytometry: Immune cell types were analysed by flow cytometry and proprietary gating algorithms at Serametrix Inc.
 MDSC were identified using the following gating strategy: PBMC/CD14$^{low/-}$/HLA-DR$^{low/-}$/CD33+/CD15+;
 Cytotoxic T-effector memory cells were identified as CD45+/CD3+/CD8+/CD45RA-/CCR7-
 Proliferating cytotoxic T-effector memory cells were identified as CD45+/CD3+/CD8+/CD45RA-/CCR7-/ki67+
- qPCR: Blood was drawn into PAXgene RNA tubes; RNA was extracted with PAXgene Blood RNA Kit (Qiagen) following manufacturer's instructions. cDNA was synthesized using SuperScript IV VILO Master Mix (Invitrogen) and ApoE and ABCA1 gene expression detected through a qPCR reaction using specific Taqman Gene Expression Assays. GUSB gene was used as an internal control.
- IHC: Formalin-fixed, paraffin-embedded slides from archival tumor biopsies were stained with ApoE antibody (ab1906, Abcam), dilution 5 μg/mL using Discovery XT Staining Module from Roche. The whole section per tumor was quantified for tumor proportion score
- All statistical analyses were conducted using R and GraphPad Prism 8; following tests were used where appropriate
 - Wilcoxon signed rank test; Mann-Whitney, Two-sample t-test

C. RGX-104 Multiple Dose Escalation Arms Study

- Patients with refractory/resistant solid tumors were enrolled in multiple dose escalation arms of RGX-104 monotherapy or in combination with checkpoint inhibitors or chemotherapy
- Prior checkpoint inhibitor therapy was allowed

RGX-104 Phase 1
Multiple escalation arm study

Treatment Arm	RGX-104 Dose	Patients Enrolled	Median Number of Prior Regimens
RGX-104 + docetaxel	80 mg BID	4	3
	80 mg (5/7)* BID	4	3
	100 mg (5/7)* BID	3	4
RGX-104 + ipilimumab	80 mg BID	7	3
	100 mg BID	4	2
	80 mg BID	4	3
RGX-104 + nivolumab	120 mg BID	5	2
	160 mg BID	7	3
RGX-104	120 mg QD	3	7
	240 mg QD	4	4
	120 mg BID	8	7
	160 mg BID	4	3
	200 mg BID	7	3
RGX-104 ovarian expansion	160 mg BID	16	5

*5 days ON, 2 days OFF

D. RGX-104 Pharmacokinetics Summary

Non-compartmental PK analysis for multiple escalation study

	RGX-104 Dose	Day	Estimated Average $AUC_{0-\infty}$ (ng-hr/mL)	SD $AUC_{0-\infty}$ (ng-hr/mL)	Average C_{max} (ng/ml)	SD C_{max} (ng/mL)
RGX-104 + docetaxel	80 mg (7/7) BID	15	13,603	11.106	951	665
	80 mg (5/7) BID*	15	7,359	2,605	785	412
	100 mg (5/7) BID*	15	7,101	1,850	618	44
RGX-104 + ipilimumab	80 mg BID	15	21,866	8,820	1,397	410
	100 mg BID	15	12,278	8,393	909	778
RGX-104 + nivolumab	80 mg BID	15	12,737	4,372	1,068	377
	120 mg BID	15	21,471	10,424	1,868	750
	160 mg BID	15	65,253	34,435	4,548	1,881
RGX-104	120 mg QD	15	12,200	3,780	1,110	722
	240 mg QD	15	35,800	31,700	3,320	1,120
	120 mg BID**	15	≥20,000	7,800	1,780	1,430
	160 mg BID**	15	≥19,280	4,120	1,810	493
	200 mg BID	15	49,313	10,032	3.028	863

*Day 15 specimens were drawn after 2 days off RGX-104
* *Estimated using a 0-8 hour sampling interval; PK parameters for all other BID cohorts are estimated using a 12 hour sampling interval

Additional PK results

- Exposure to RGX-104 reached steady state on Cycle 1 Day 15
- Half life ranged from 6-8 hours
- Partial responses were observed at steady state exposure of 10,000 to 15,000 ng*h/mL RGX-104 (monotherapy and in combination)

E. RGX-104 Treatment Effects on ApoE Induction

RGX-104 activates ApoE expression in circulating immune cells of treated patients

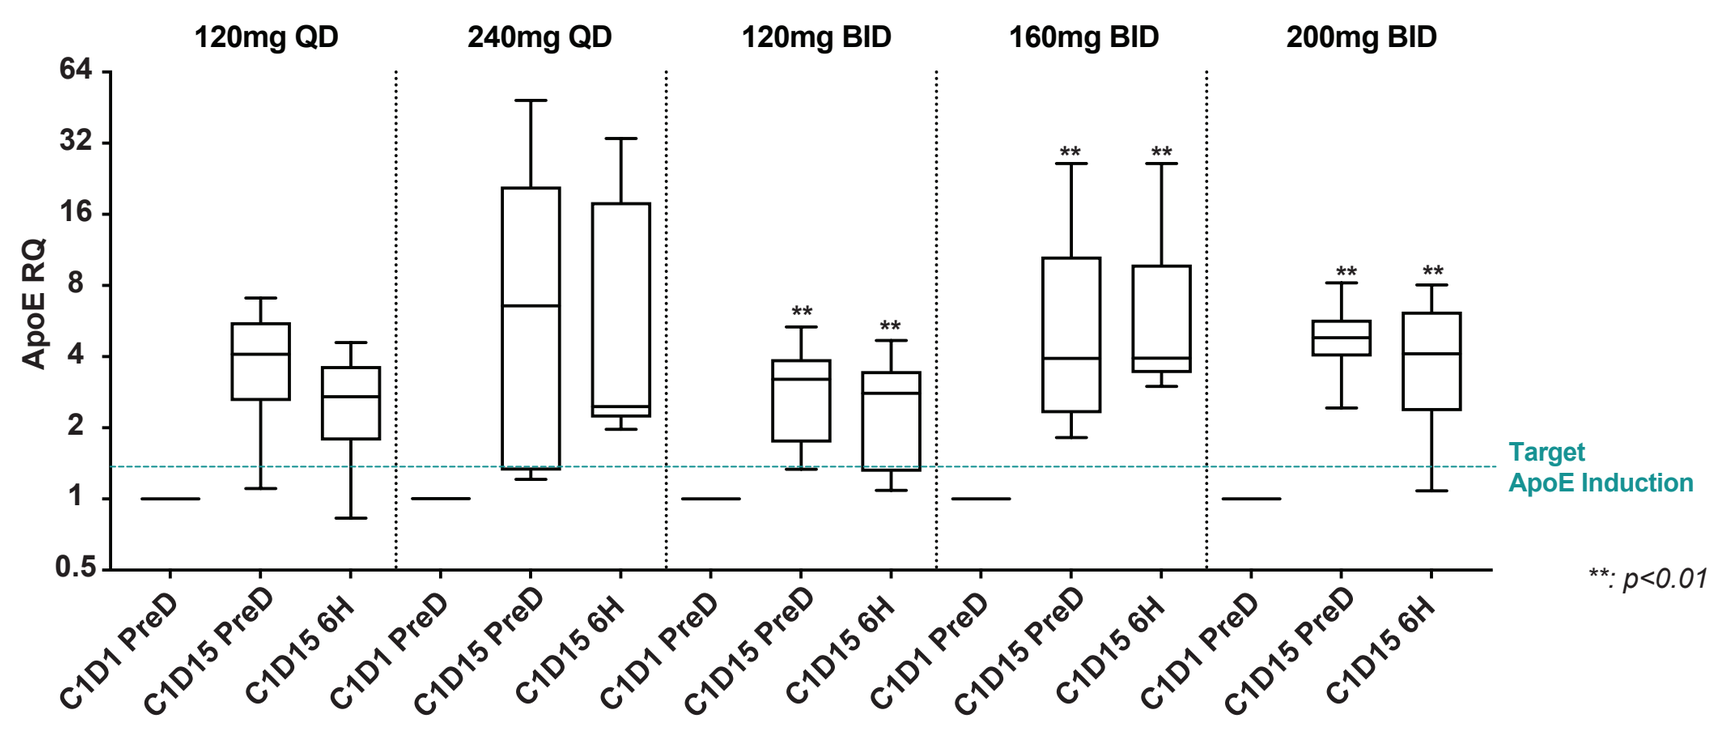
120mg QD
240mg QD
120mg BID
160mg BID
200mg BID
ApoE RQ
Target ApoE Induction
**: p<0.01
C1D1 PreD
C1D15 PreD
C1D15 6H

- Expression of LXR target genes, ApoE and ABCA1, was used as pharmacodynamic readout of target engagement by RGX-104 using a qPCR method that was developed and qualified to monitor gene expression in peripheral whole blood specimens
- Whole blood drawn at pre-dose time points on Days 1 and 15 of cycle 1 was used to assess RGX-104 activity reflecting gene induction at daily trough levels of RGX-104
- A general increase in ApoE gene induction in circulating immune cells was noted with increasing daily dose
- Sustained gene induction was better with BID dosing compared to that from QD dosing: steady state induction at pre-dose time points is statistically significant in BID cohorts
- In preclinical models, 1.5- to 2.5-fold induction of ApoE is sufficient to drive maximal anti-tumor effects
- Target pharmacodynamic induction was achieved at doses ≥ 120mg BID; gene induction at 160mg BID was comparable to that at 200mg BID
- Similarly, induction of ABCA1, an LXR target gene with rapid induction kinetics, is detectable starting at 4-6h after dose on cycle 1 day 1, suggesting immediate target engagement by RGX-104 (data not shown)

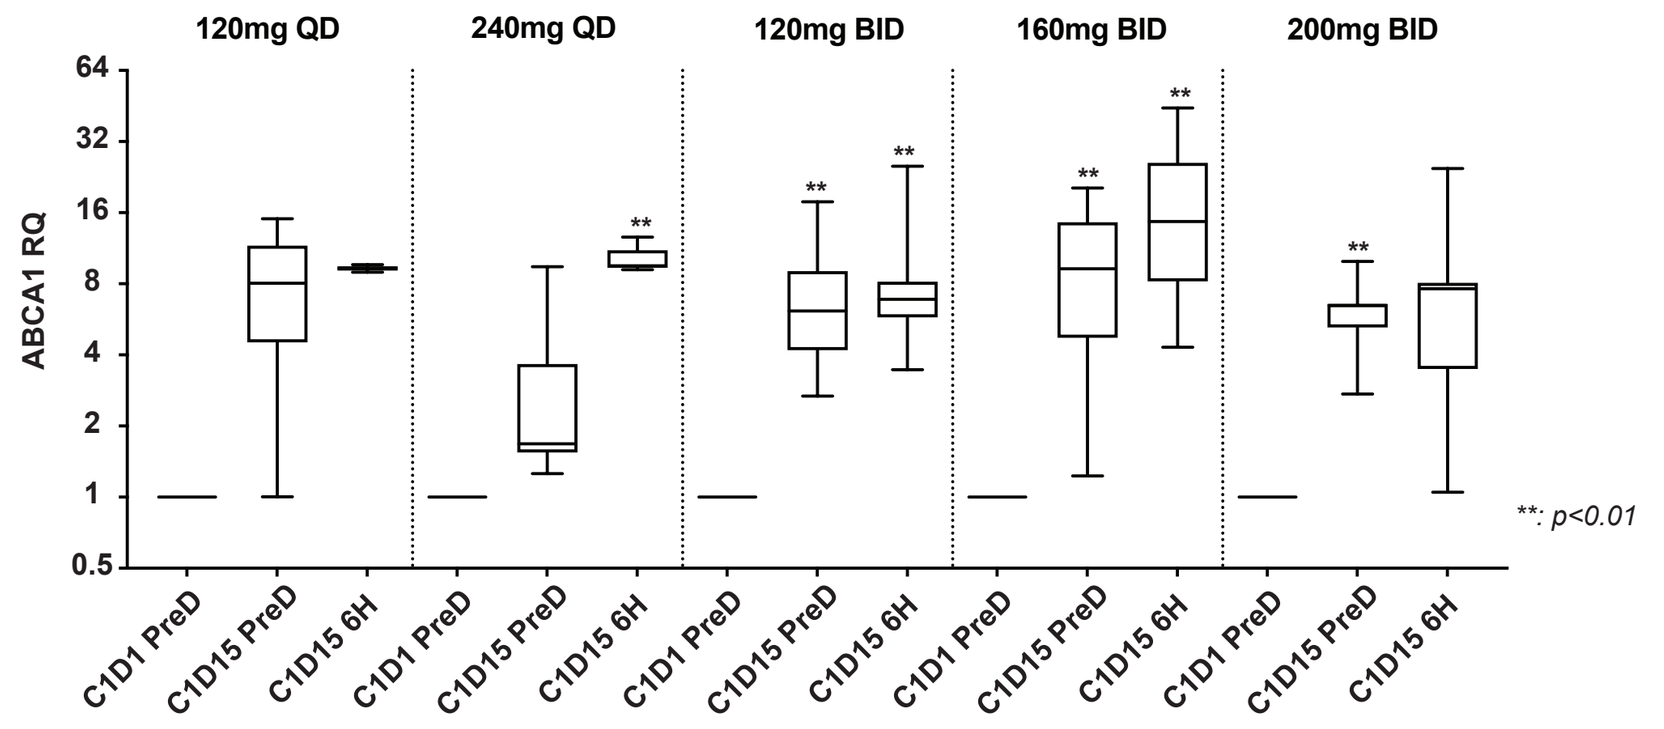
120mg QD
240mg QD
120mg BID
160mg BID
200mg BID
ABCA1 RQ
**: p<0.01
C1D1 PreD
C1D15 PreD
C1D15 6H

E.1 ApoE Induction Correlates with MDSC Depletion

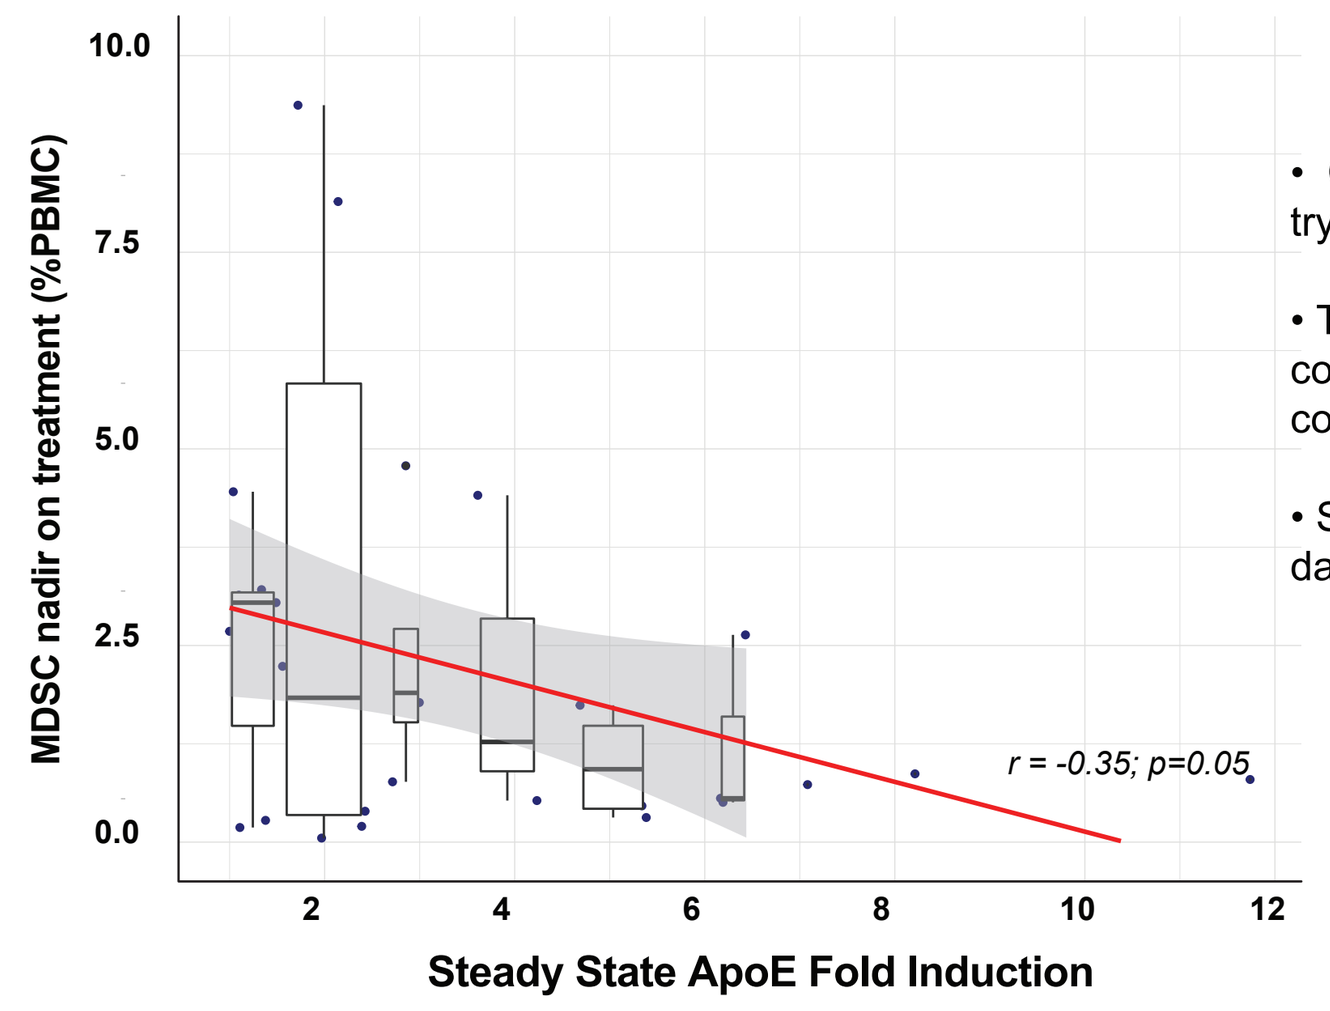
MDSC nadir on treatment (%PBMC)
Steady State ApoE Fold Induction
r = -0.35; p=0.05

- Circulating MDSC abundance was monitored by flow cytometry during the first two cycles of treatment with RGX-104
- The absolute minimum level of MDSC (nadir) on treatment correlated negatively with the extent of ApoE gene induction, consistent with ApoE-mediated MDSC depletion with RGX-104
- Steady state gene induction measured by qPCR on cycle 1 day 15 expressed as RQ were used for correlative analyses

F. RGX-104 Monotherapy Activity and Correlation with MDSC Depletion

RGX-104 monotherapy clinical activity

Response in patients receiving ≥160mg BID RGX-104

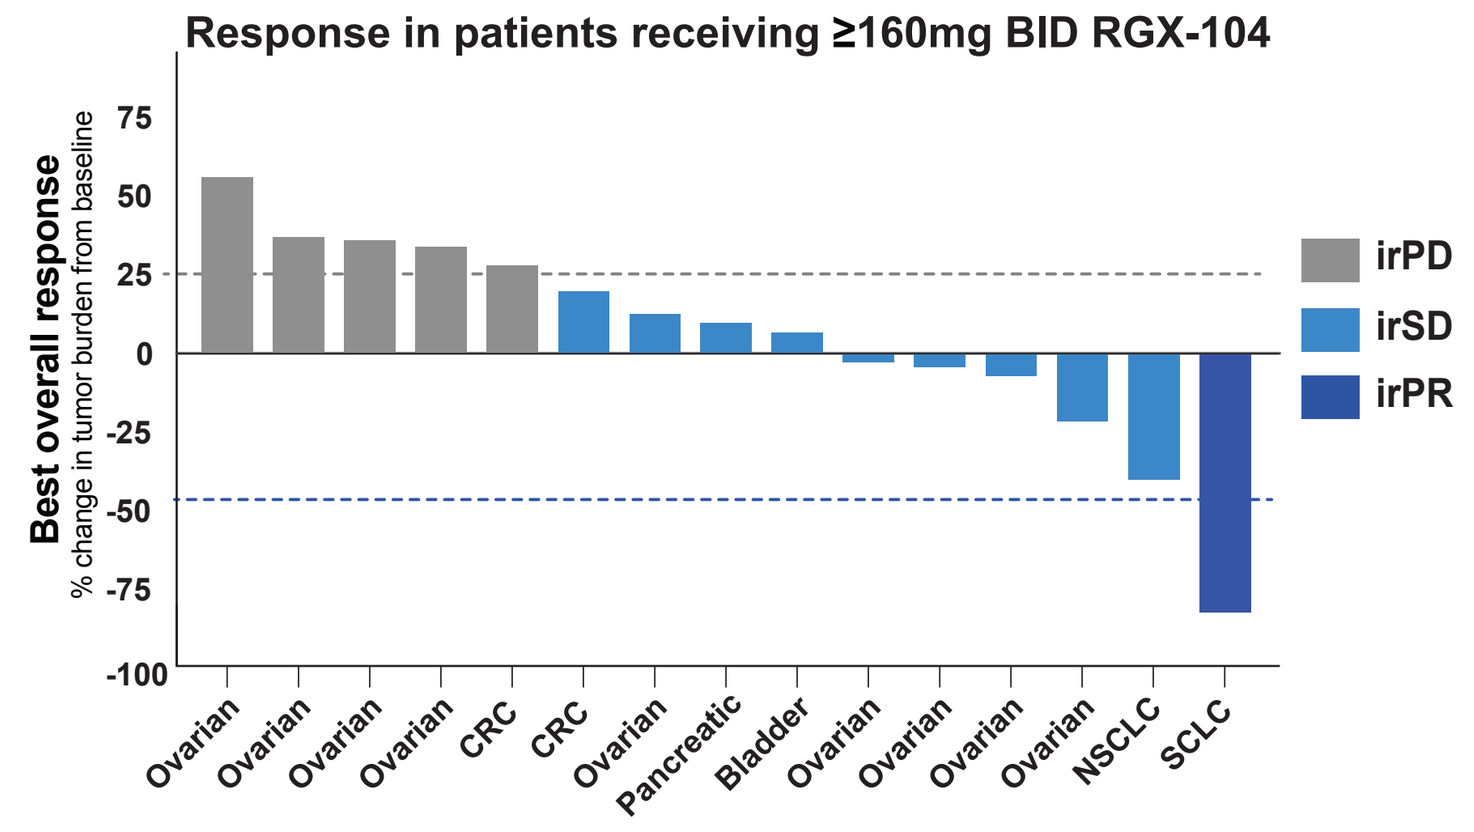
Best overall response
% change in tumor burden from baseline
irPD
irSD
irPR
Ovarian
CRC
Pancreatic
Bladder
NSCLC
SCLC

- 15 patients with relapsed/refractory solid tumors treated with RGX-104 at dose ≥160mg BID were evaluable for response: n=10 (160mg BID); n=5 (200mg BID)
- Of these, 6 (40%) patients had tumor regressions as determined by immune related response criteria (irRC): 1 partial response and 5 stable disease

RGX-104 clinical response is associated with MDSC depletion

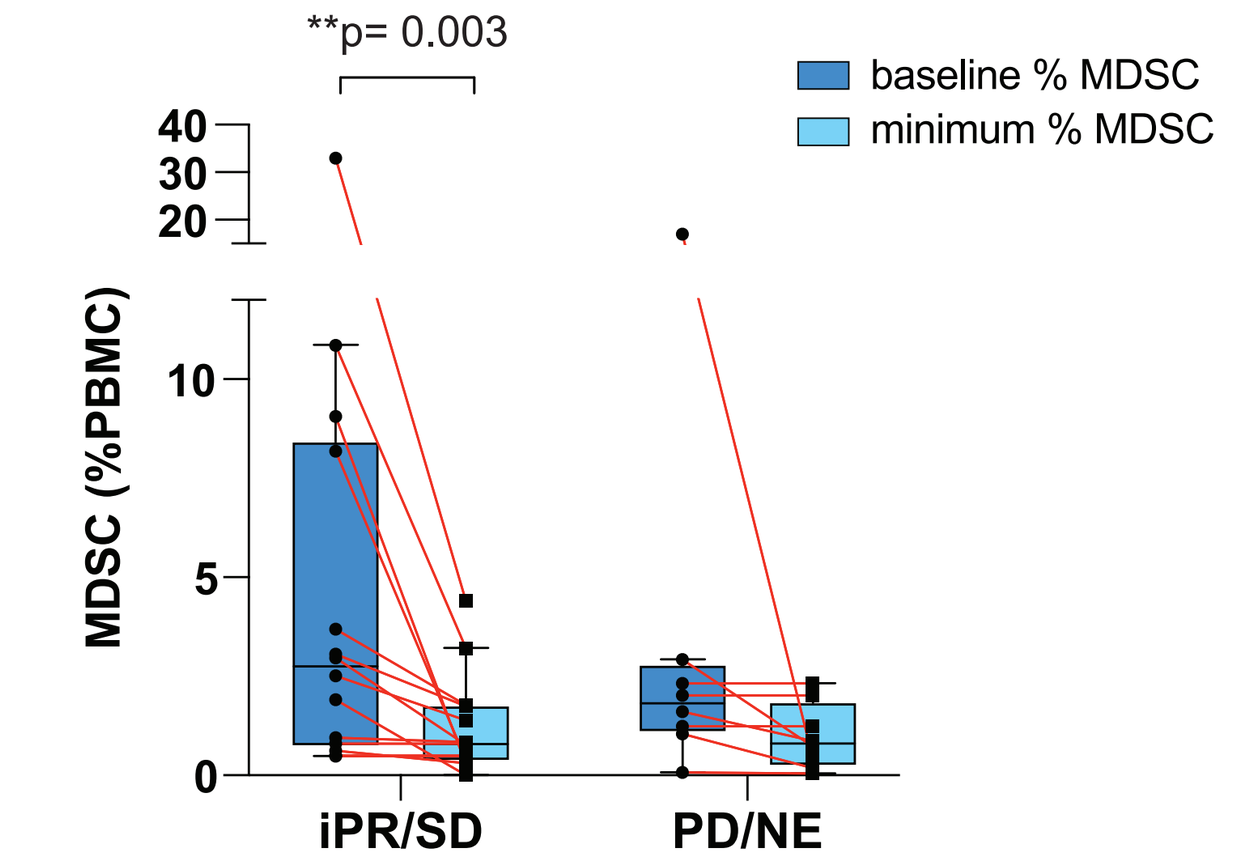
**p= 0.003
baseline % MDSC
minimum % MDSC
MDSC (%PBMC)
iPR/SD
PD/NE

- Patients with SD or PR had a significant depletion of MDSC on treatment

Patients with clinical response have lower baseline (pre-treatment) intratumoral ApoE

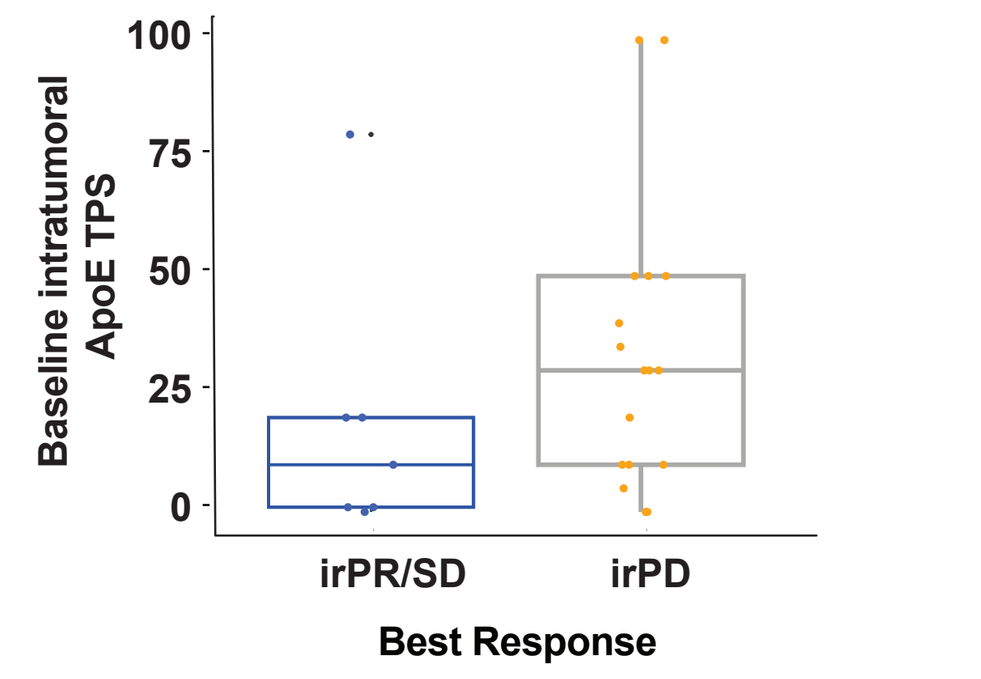
Baseline intratumoral ApoE TPS
irPR/SD
irPD
Best Response

- Consistent with RGX-104 MOA, patients with low baseline levels of tumoral ApoE experienced greater clinical benefit with RGX-104 therapy

G. Clinical Activity of RGX-104 Combination Regimens

Tumor response after treatment with RGX-104 in combination with checkpoint inhibitors (CPI) or chemotherapy

Treatment Arm	Tumor Types Enrolled	Longest Response	irPR	irSD	irPD	Disease Control Rate
RGX-104 + docetaxel	adrenal, bladder, endometrial, neuroendocrine, ovarian, melanoma, SCCHN	>11 months (ongoing PR)	2	4	3	66.6%
RGX-104 + ipilimumab	anal, CRC, endometrial, NSCLC, prostate, SCLC	>6 months (ongoing PR)	1	2	4	42.8%
RGX-104 + nivolumab	bladder, breast, endometrial, melanoma, NSCLC, prostate, renal, SCLC	>12 months (ongoing PR)	1	4	5	50.0%

Response Rates in Patients Previously Treated with CPI (n=14)

irPR	irSD	irPD
4 (28%)	5 (36%)	5 (36%)

Best response in all evaluable patients previously treated with a CPI
Data from RGX-104 combination dose escalation cohorts

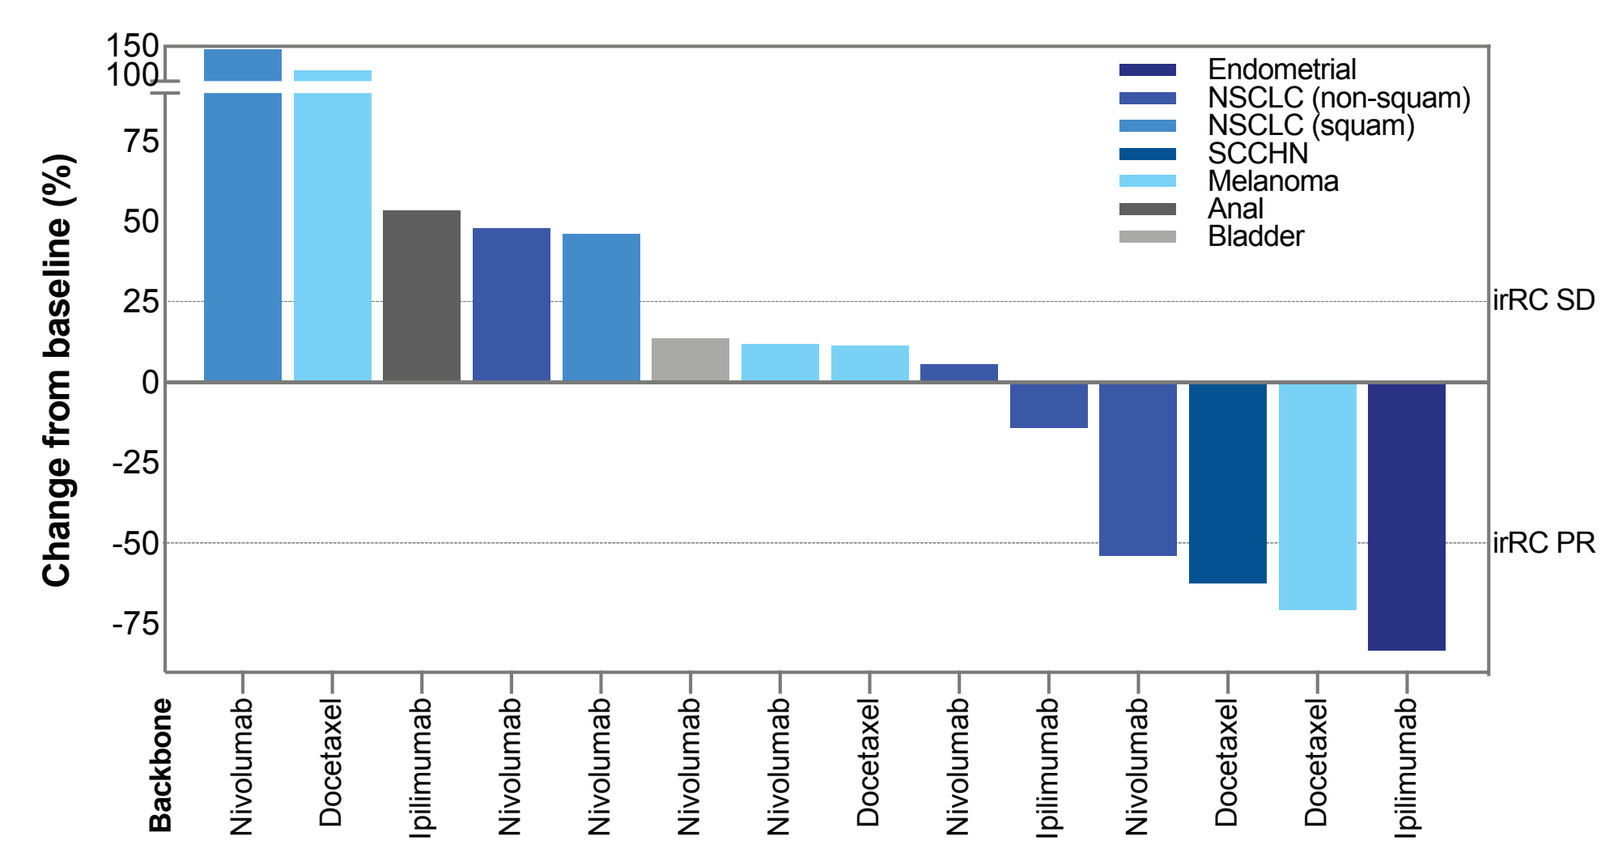
Change from baseline (%)
Endometrial
NSCLC (non-squam)
NSCLC (squam)
SCCHN
Melanoma
Anal
Bladder
irRC SD
irRC PR
Backbone
Nivolumab
Docetaxel
Ipilimumab

- 26 patients with relapsed/refractory solid tumors treated with RGX-104 in combination with docetaxel, ipilimumab or nivolumab were evaluable for response with at least one follow up scan
- 14 of these patients had previously progressed on check point inhibitor (CPI) therapy; among those CPI-treated patients the overall response rate was 28.6% with a disease control rate of 64.3% (4 patients with irPR and 5 patients with irSD), with durable clinical activity >12 months observed
- Durable PRs were observed in PD-L1 low/negative tumors who were previously treated with CPI:
 - NSCLC (PD-L1 TPS: 1%); PD as best response to prior nivolumab; PR>12 months (ongoing)
 - Endometrial (MSS, PD-L1 TPS: 0%); SD as best response to prior durvalamab; PR>6 months (ongoing)

G.1 RGX-104 Effects on MDSC Abundance and T Cell Activation

MDSC depletion is evident during the first cycle of treatment

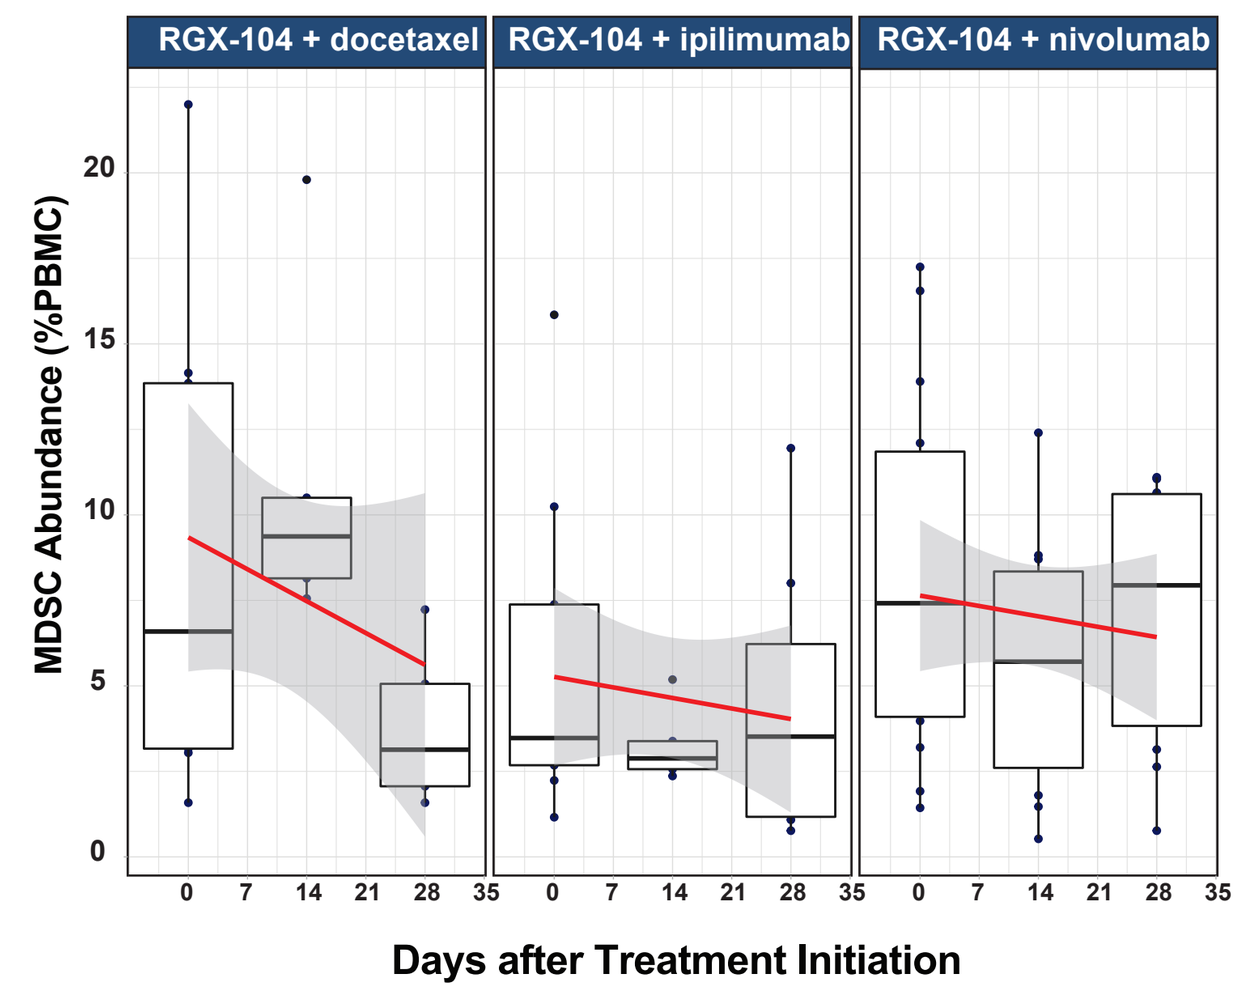
RGX-104 + docetaxel
RGX-104 + ipilimumab
RGX-104 + nivolumab
MDSC Abundance (%PBMC)
Days after Treatment Initiation

Increase in proliferating CD8 T-effector memory cells reaches a peak during Cycle 2

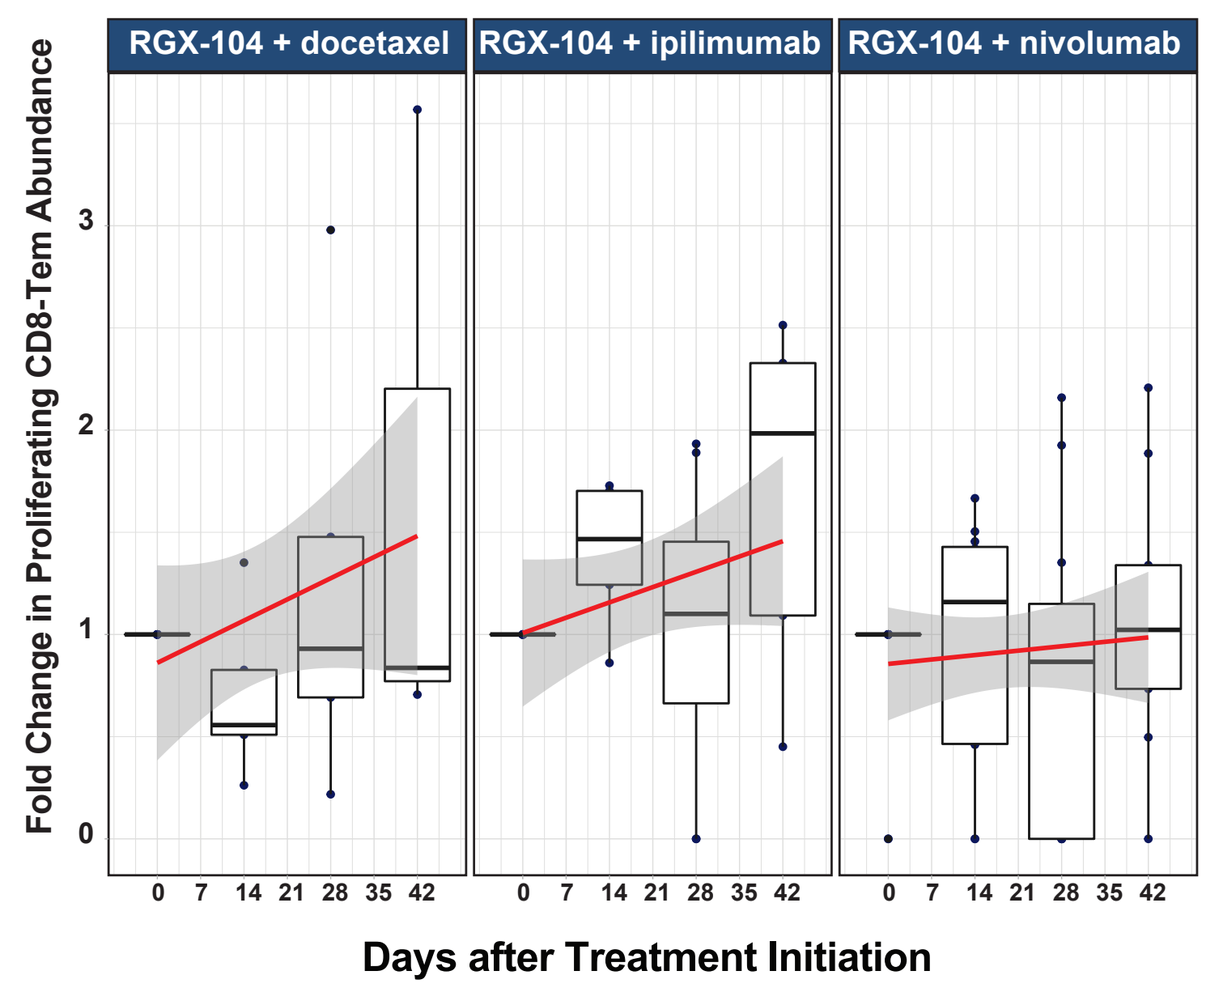
RGX-104 + docetaxel
RGX-104 + ipilimumab
RGX-104 + nivolumab
Fold Change in Proliferating CD8-Tem Abundance
Days after Treatment Initiation

T cell activation is associated with clinical activity across all RGX-104 combination cohorts

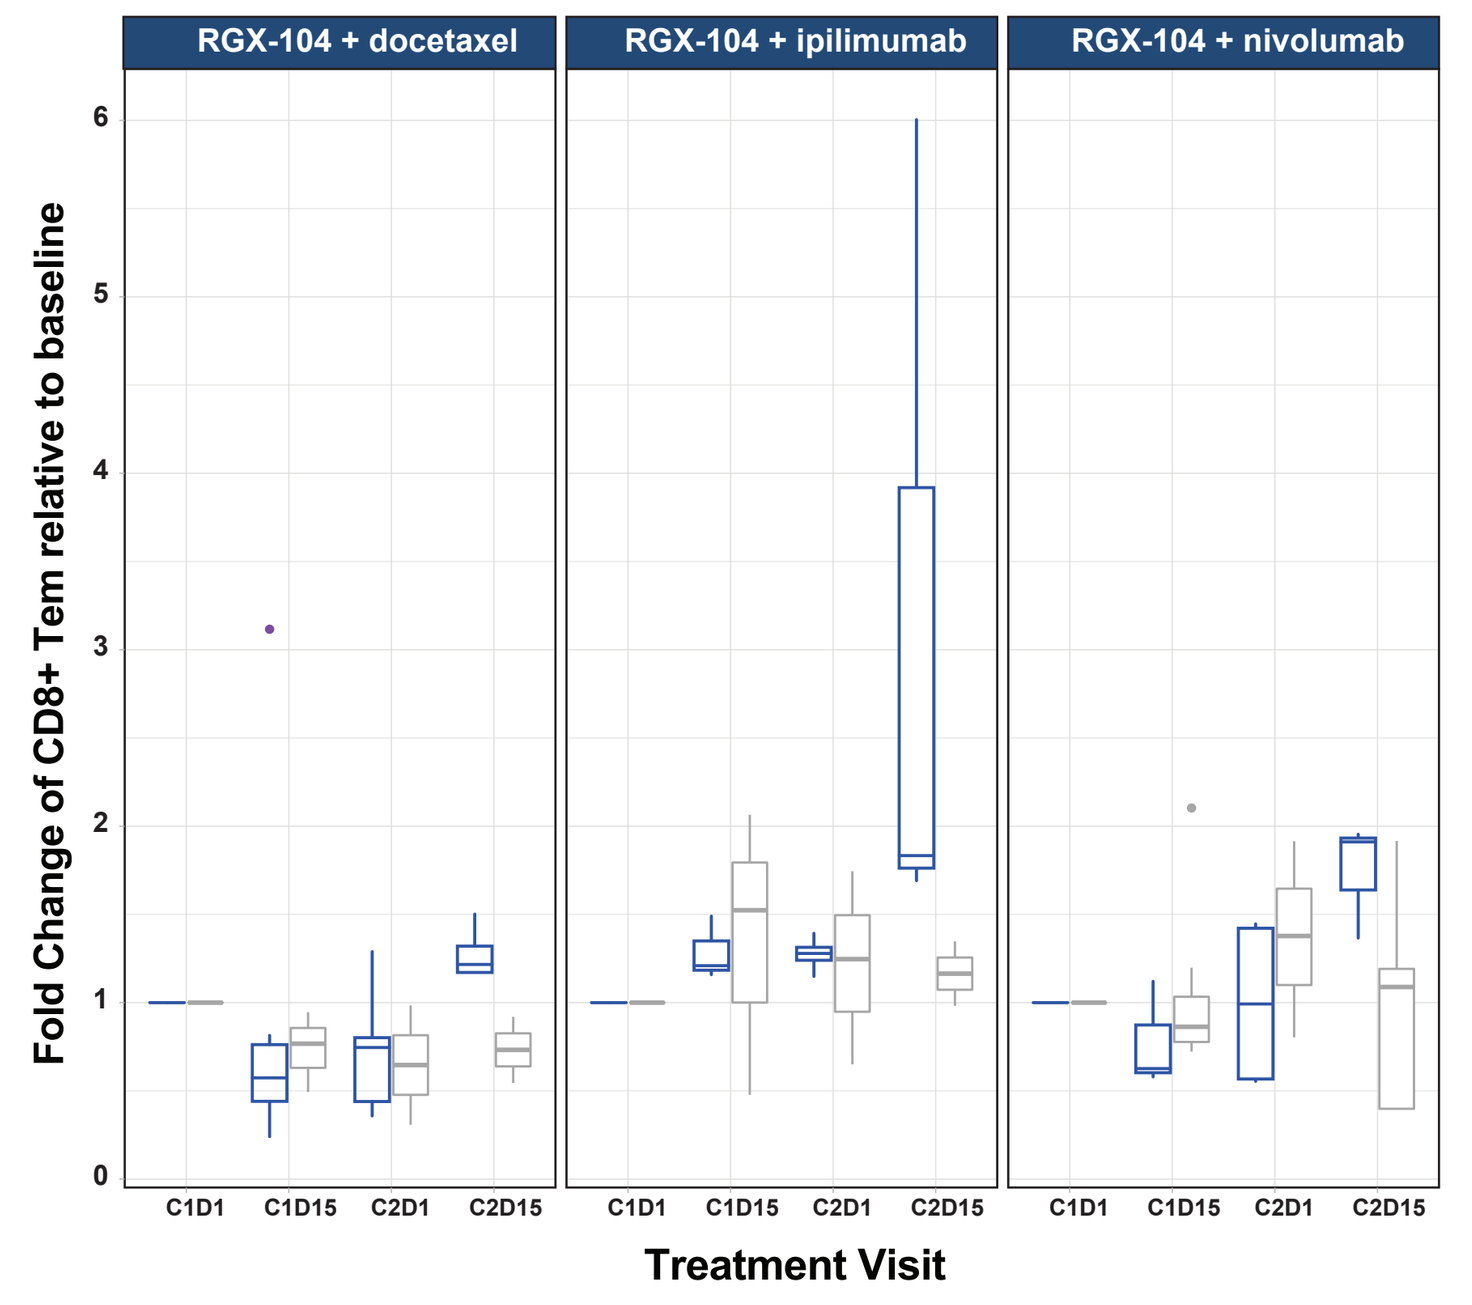
RGX-104 + docetaxel
RGX-104 + ipilimumab
RGX-104 + nivolumab
Fold Change of CD8+ Tem relative to baseline
C1D1
C1D15
C2D1
C2D15
Treatment Visit
Best Response
irPR/SD
irPD

- Immune cell profiling in patients enrolled in combination cohorts demonstrated robust reduction of circulating MDSC during the first cycle of treatment
- T cell activation was evident as demonstrated by an increase in proliferating CD8 T-effector memory (Tem) cells (as a percentage of total CD8 Tem) relative to baseline
- T cell responses were evident after the first cycle of treatment and generally occured after peak MDSC depletion was achieved, consistent with reversal of MDSC-mediated T cell suppression with RGX-104 treatment
- Induction of T cell responses was associated with clinical activity, with more pronounced effects on total CD8-Tem in patients with irPR/SD as compared to that in patients PD at the cycle 2 day 15 visit

H. RGX-104 Mediated Cytokine Effects

RGX-104 dependent induction of IFN-γ and IL-5 and their association with treatment outcome

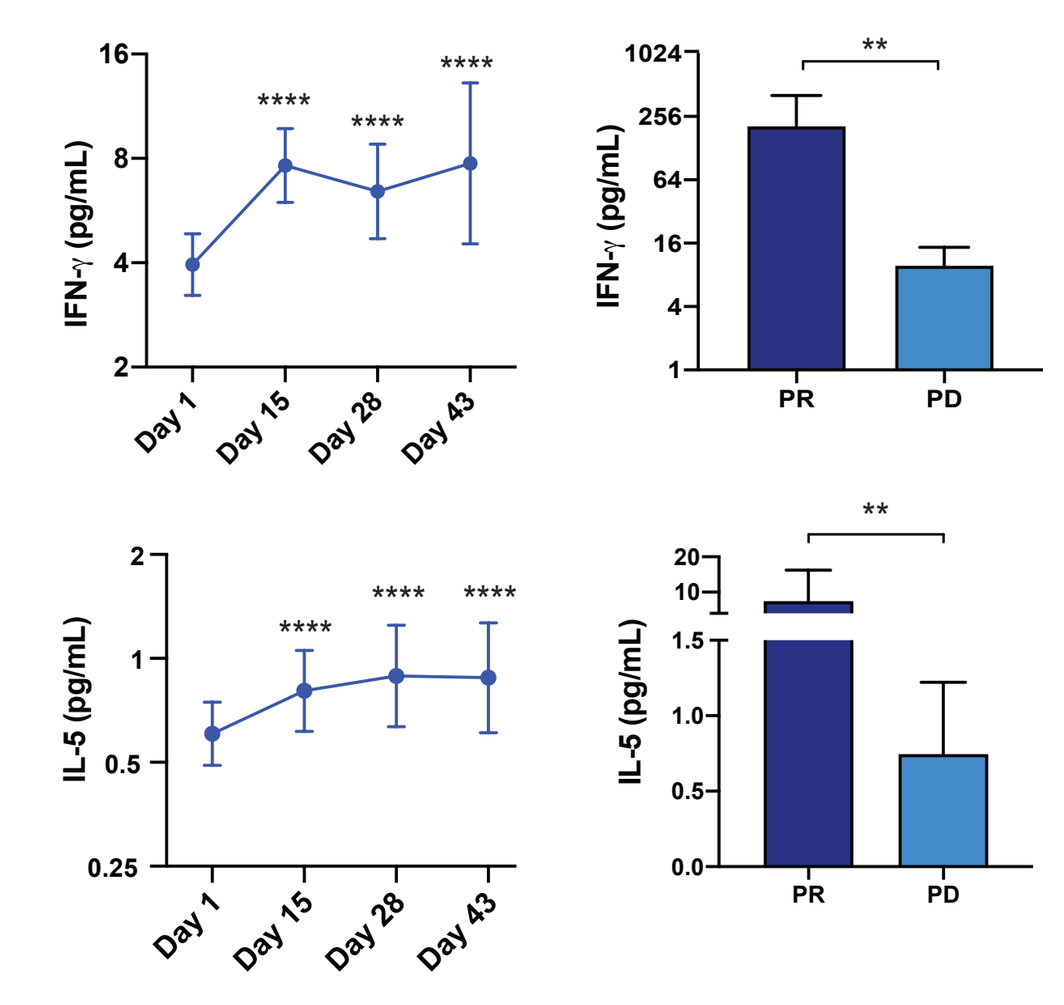
IFN-γ (pg/mL)
IL-5 (pg/mL)
Day 1
Day 15
Day 28
Day 43
PR
PD
** p< 0.01
**** p< 0.0001

- IFN-γ is a Th1 cytokine associated with effector T cell responses and is increased in the serum of patients with evaluable samples in single agent and combination cohorts, reaching peak levels concomitant with increases in activated CD8 T cell effector populations
- IL-5 is a Th2 cytokine known to be induced in myeloid cells by LXR agonist treatment (5) and was found to rapidly increase in the serum of treated patients with a time course concomitant with RGX-104-mediated MDSC depletion
- Higher levels of IL-5 and IFN-γ were associated with better treatment outcome across cohorts

I. Association Between ApoE and PD-L1 Expression in Patient Tumors

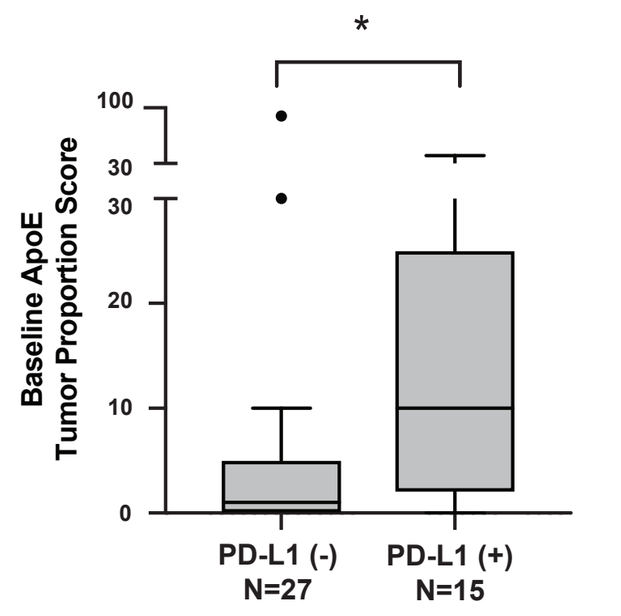
Baseline ApoE Tumor Proportion Score
PD-L1 (-) N=27
PD-L1 (+) N=15

- Patients with tumors that are PD-L1 negative are more likely to have low levels of ApoE expression at baseline thereby making them better candidates for treatment with RGX-104
- An expansion cohort of treatment naive NSCLC patients whose tumors have PD-L1 <1% is currently being enrolled for treatment with RGX-104 in combination with pembrolizumab, carboplatin and pemetrexed

J. Case Study Subject 016-5202: Clinical response in CPI refractory patient associated with RGX-104 mediated immune activation

- 62 y old female with SCCHN of the hypopharynx, previously treated with and progressed on chemotherapy, cetuximab and nivolumab
- CPI refractory (best response to prior nivolumab therapy: progressive disease)
- Baseline ApoE Tumor Proportion Score: 2%
- On study treatment: RGX-104 80mg (5/7) BID + docetaxel
- Partial Response (irPR) on therapy with 62% reduction in tumor burden including large (>7cm) primary lesion and hepatic metastases

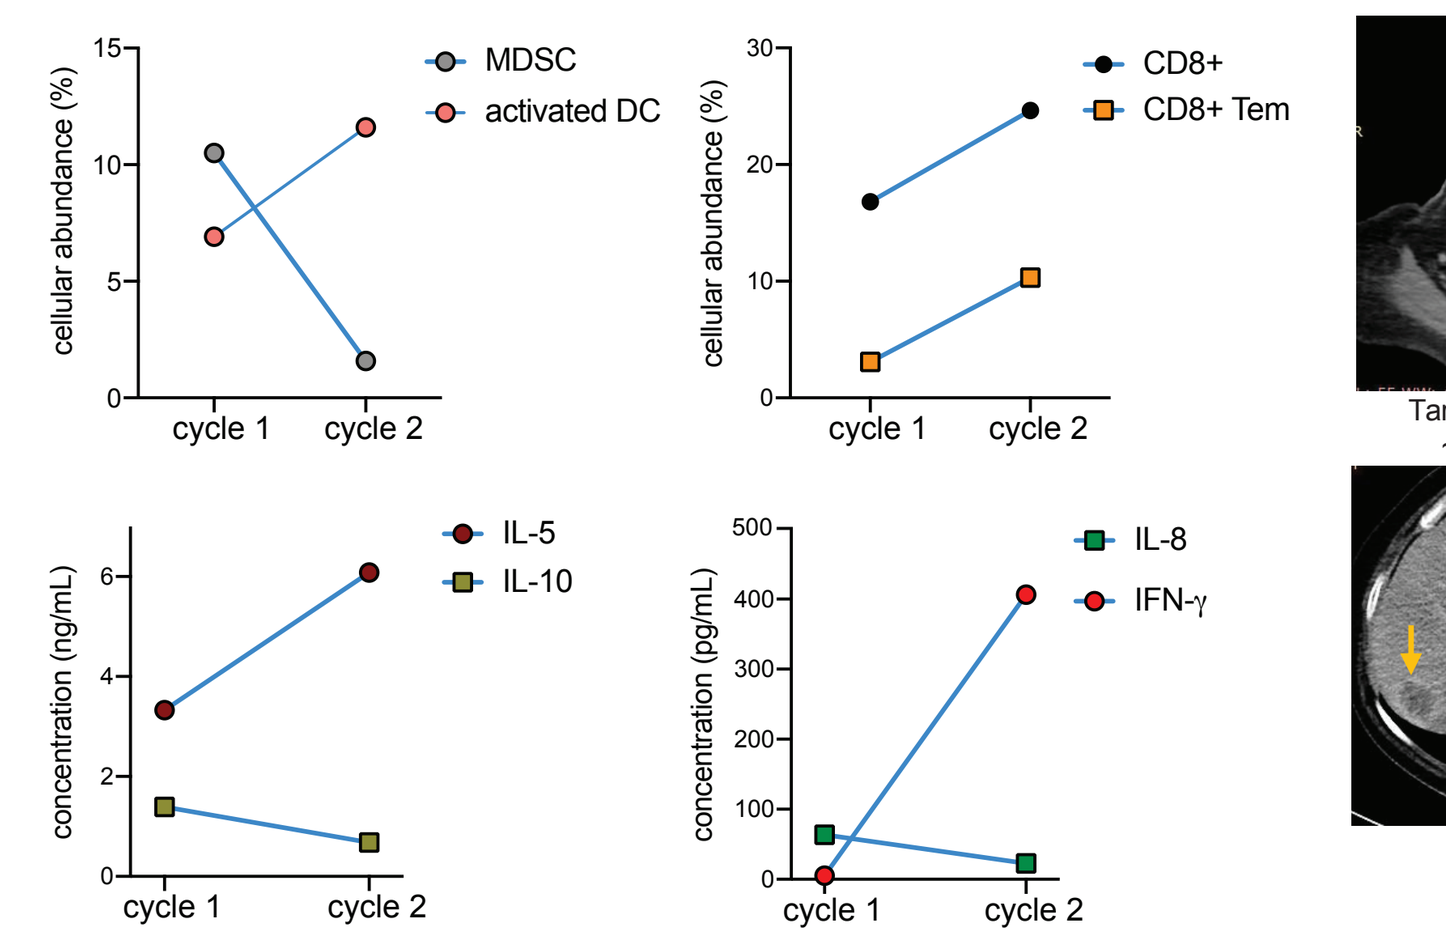
MDSC
activated DC
CD8+
CD8+ Tem
IL-5
IL-10
IL-8
IFN-γ
cellular abundance (%)
concentration (ng/mL)
concentration (pg/mL)
cycle 1
cycle 2

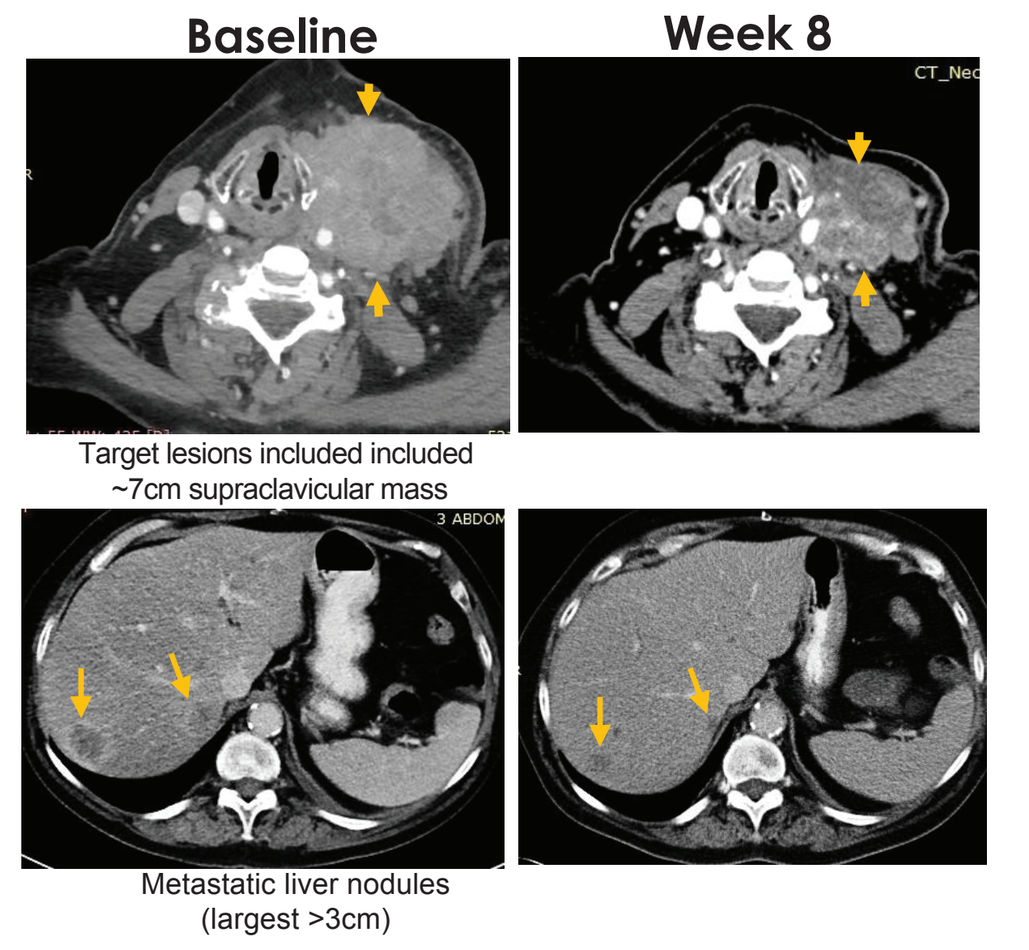
Baseline
Week 8

Target lesions included included ~7cm supraclavicular mass

Metastatic liver nodules (largest >3cm)

- Decrease in MDSC abundance was associated with an increase in activated dendritic cells and an increase in total CD8+ T cells and CD8+ T effector memory cells
- Cytokine analysis revealed an increase in the serum levels of IL-5 and IFN-γ and a decrease in the levels of IL-10 and IL-8 after treatment with RGX-104 + docetaxel

K. Conclusions

- In all cohorts analyzed, a temporal dynamic of RGX-104-mediated gene induction was observed; BID dosing was better than QD dosing in maintaining drug-induced expression of target gene ApoE at steady state
- MDSC depletion was observed with increasing RGX-104-mediated induction of ApoE and was associated with clinical response; patients with irPR/SD exhibited a more significant reduction in circulating MDSC compared to that in patients with progressive disease, consistent with the MOA of RGX-104
- MDSC depletion correlated with an increase in circulating cytotoxic CD8+ T-effector memory cells that was associated with clinical response across cohorts
- Cytokines relevant to effector CD8+ T cell responses and LXR signalling in myeloid cells were increased in the serum of patients, and associated with clinical activity
- Patients with lower levels of baseline intratumoral ApoE had favorable treatment outcomes; these patients are also more likely to have low levels of tumoral PD-L1, providing biological rationale for RGX-104 treatment in PD-L1 low/negative patients
- In addition to monotherapy anti-tumor activity in several tumor types (NSCLC, SCLC, and ovarian cancer), durable clinical activity was observed in patients across combination cohorts in CPI resistant/refractory patients including those harboring PD-L1 low/negative tumors. This demonstrates RGX-104-mediated reversal of drug resistance via MDSC depletion
- An expansion cohort in treatment naive NSCLC patients with PD-L1<1% is currently enrolling to assess efficacy of RGX-104 in combination with pembrolizumab, carboplatin and pemetrexed
- An expansion cohort in 2L SCLC/HG-NET patients is currently enrolling to assess efficacy of RGX-104 in combination with docetaxel

RGX-104 Phase 1b/2 cohorts

- **1L non-squamous NSCLC$^{(PD-L1 < 1\%)}$** RGX-104 + Pembrolizumab + Carboplatin/Pemetrexed
- **2L SCLC/HG-NET** RGX-104 + Docetaxel

References

1. Pencheva N et al. Cell. 2012 Nov; 151(5):1068-82
2. Pencheva N et al. Cell. 2014 Feb; 156(5):986-1001
3. Tavazoie M et al. Cell. 2018 Feb; 172, 825–840
4. Lee and Tontonoz, Atherosclerosis. 2015; 242:29-36
5. Chen Y et al. J Biol Chem. 2012 Dec 21; 287(52): 43340–43350

Acknowledgements

The authors thank all patients and their families and site personnel
The authors would also like to acknowledge staff at Precision Oncology, Precision for Medicine, Inventiv/Syneos, Serametrix and Cancer Genetics Inc. for their support with the clinical trial and with various clinical sample analyses and assays

For further information, please contact: foster.gonsalves@rgenix.com